Effects of the Mitochondria-Targeted Drug Elamipretide on Leakage-Independent Vision Loss in Fellow Eyes with Neovascular AMD in the ReCLAIM Study

358
A0235

Priyatham S. Mettu, Michael J. Allingham, and Scott W. Cousins
Ophthalmology / Duke Eye Center, Duke University School of Medicine, Durham, NC

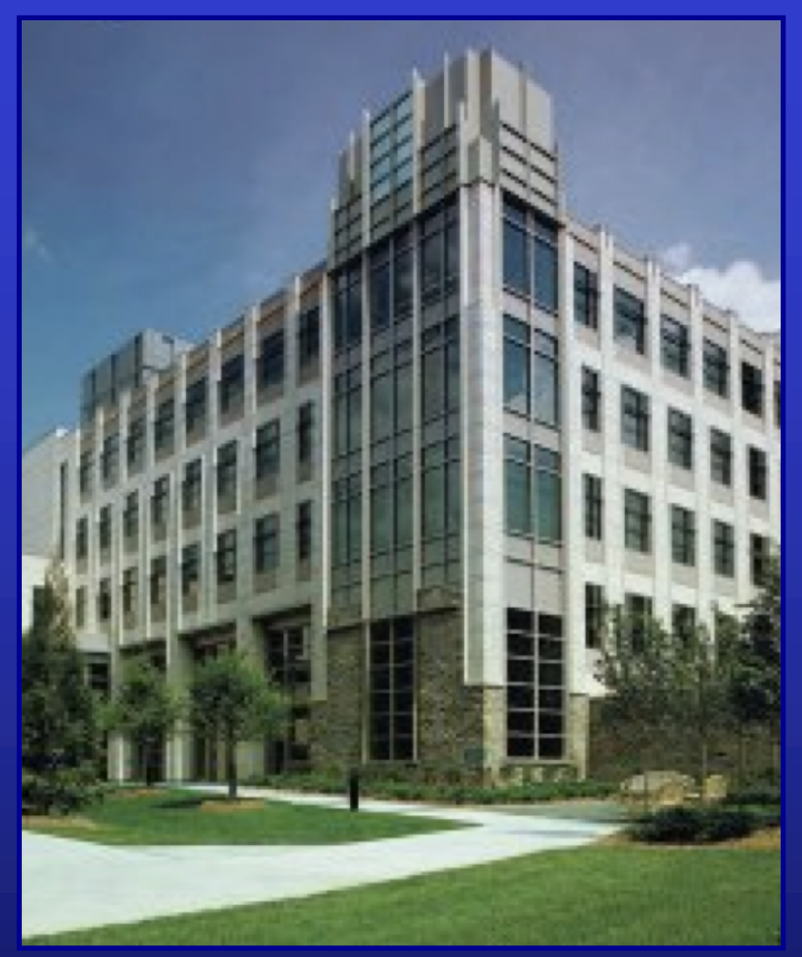

ABSTRACT

Purpose: Assess the effects of the mitochondria-targeted drug elamipretide on leakage-independent vision loss in fellow, non-study eyes with neovascular age-related macular degeneration (NV AMD) in the ReCLAIM Study, an open-label, phase 1 clinical trial to evaluate safety, tolerability, and efficacy of elamipretide in dry AMD.

Methods: Eligible participants had a single designated study eye with dry AMD (either high-risk drusen or noncentral geographic atrophy); the fellow, non-study eye could have any stage of dry or NV AMD disease. All participants received daily subcutaneous elamipretide (40 mg) for 24 weeks, and study assessments were performed for both study eye and fellow, non-study eye of participants every four weeks, with outcomes assessed at week 24. The present analysis includes only fellow, non-study eyes with either quiescent NV AMD (i.e. no fluid, leakage, or hemorrhage and stable maintenance anti-VEGF treatment regimen) or stable inactive NV AMD. Visual function studies included standard luminance best-corrected visual acuity (BCVA) and low luminance visual acuity (LLVA), defined as BCVA through a log 2 neutral density filter.

Results: Of 40 participants who received study drug, a total of thirteen had fellow, non-study eyes with either quiescent NV AMD (n= 8) or stable inactive NV AMD (n= 5), with all eyes free from exudation and hemorrhage. The mean change in standard BCVA in fellow eyes with NV AMD was +6.3 $\pm$ 5.0 letters ($p < 0.001$) (baseline mean BCVA: 53.4 $\pm$ 21.9 letters, range: 19-88 letters;), with ≥ 6-letter BCVA gain in 54% of fellow eyes and ≥ 10-letter BCVA gain in 31% of fellow eyes. The mean change in LLVA in fellow eyes with NV AMD was +6.4 $\pm$ 4.8 letters ($p < 0.001$) (baseline mean LLVA: 35.2 $\pm$ 22.9 letters, range: 6-74 letters), with ≥ 6-letter LLVA gain in 54% of fellow eyes and ≥ 10-letter LLVA gain in 31% of fellow eyes.

Conclusions: Subcutaneous administration of elamipretide, a mitochondria-targeted drug, was associated with improved vision in fellow, non-study eyes with NV AMD in a phase 1 clinical trial of elamipretide for dry AMD. Further study is warranted to understand the biology of retinal mitochondrial dysfunction in NV AMD and to assess the efficacy of elamipretide for treatment of leakage-independent vision loss in NV AMD.

BACKGROUND

MOA: **Elamipretide, tetrapeptide drug, targets mitochondria, binding damaged cardiolipin and reversing mitochondrial dysfunction**

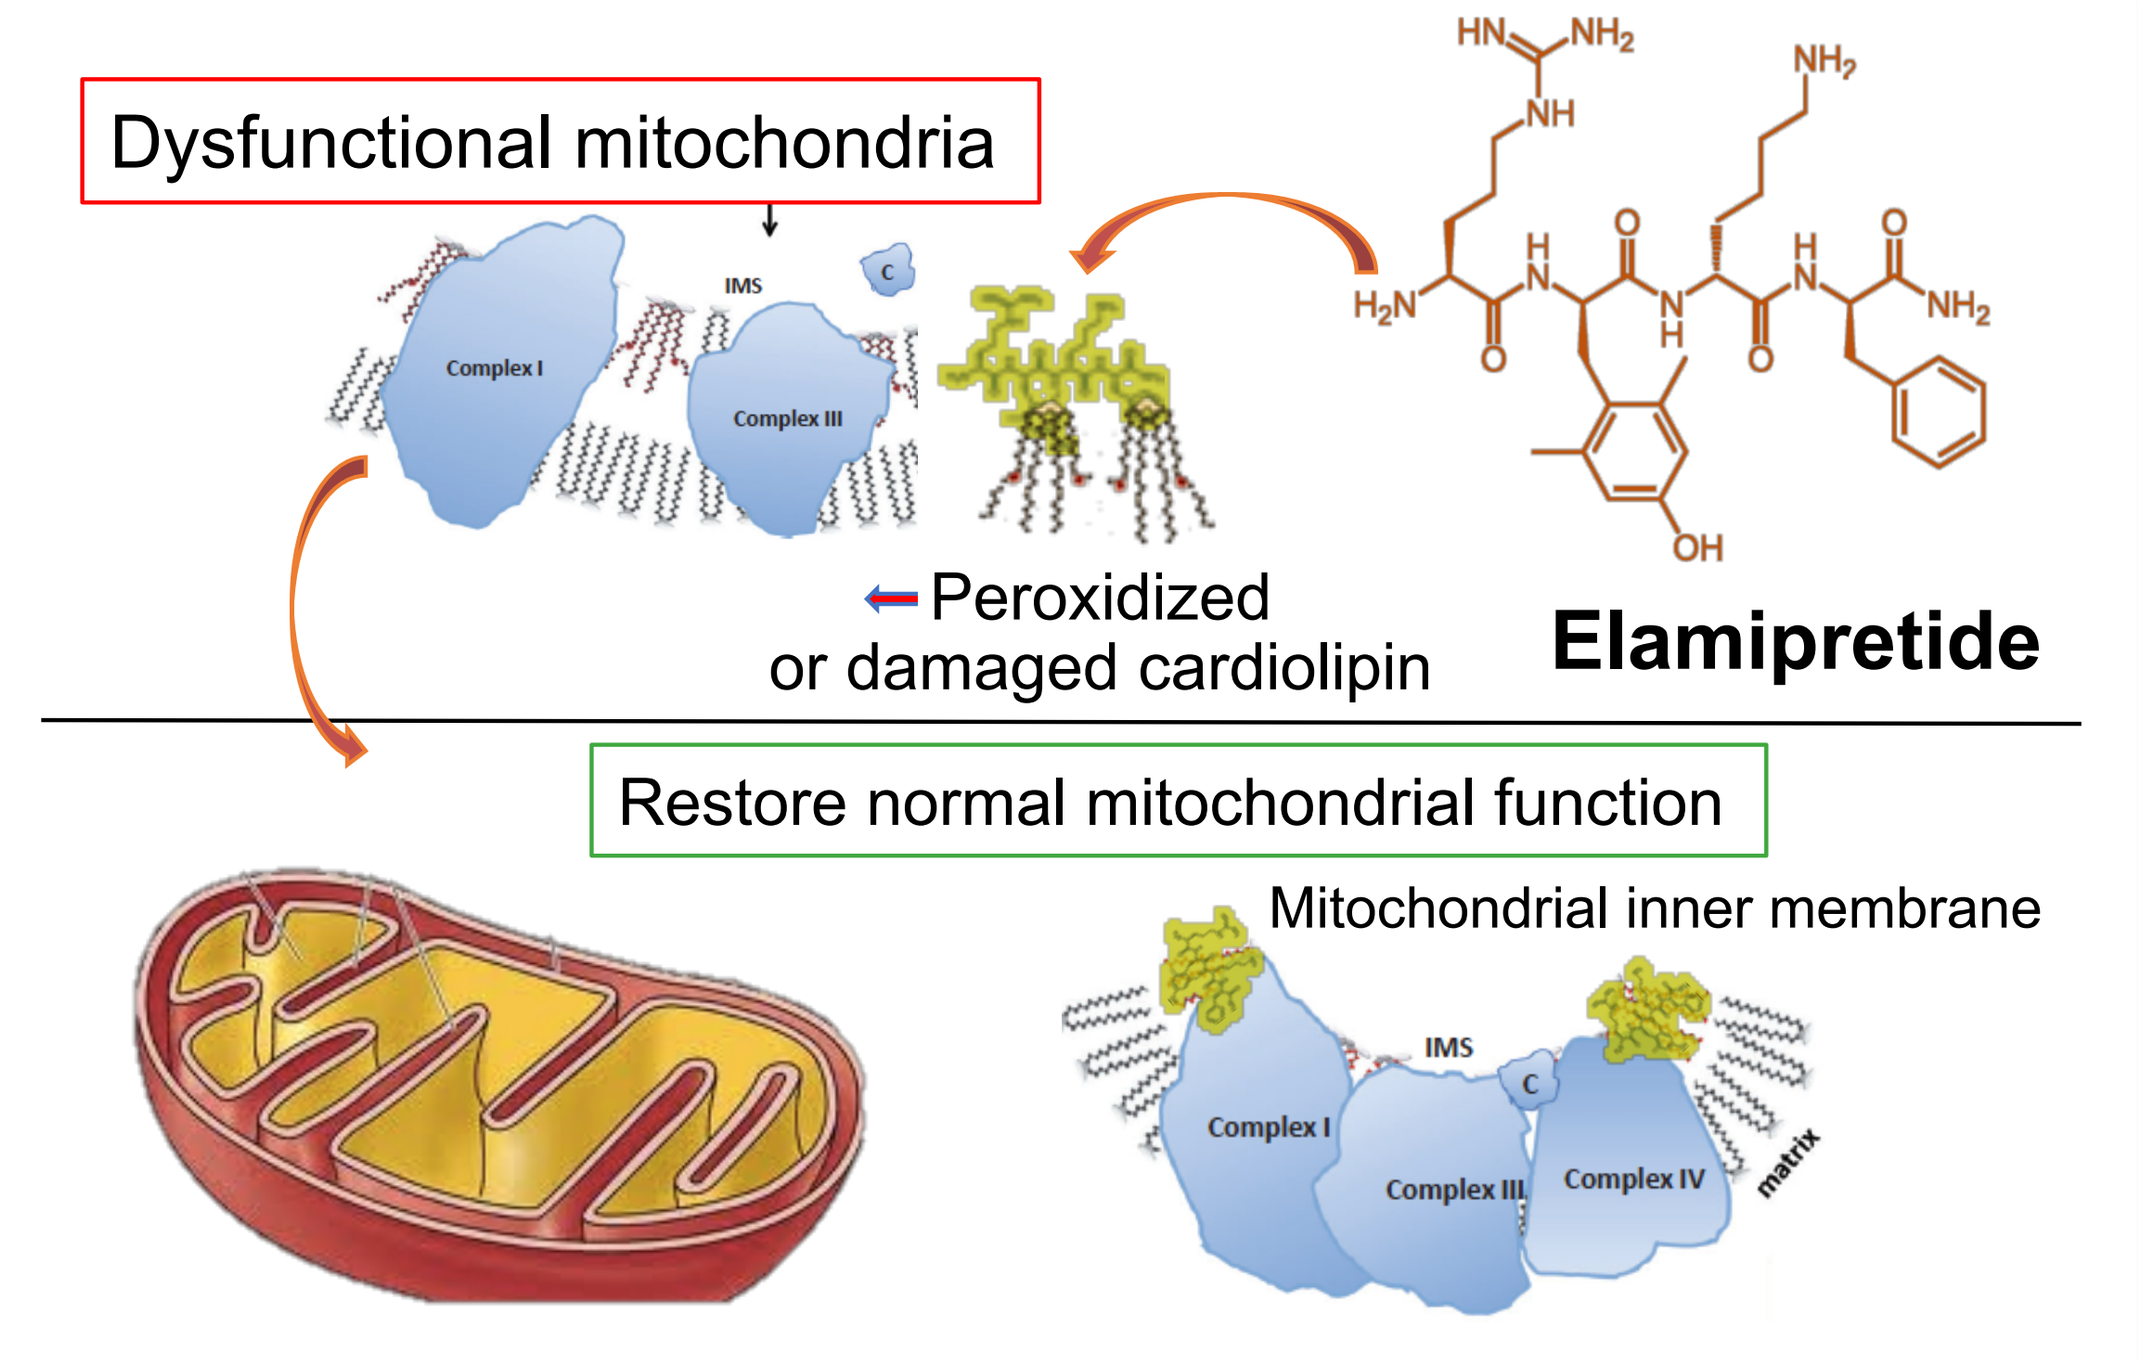
Dysfunctional mitochondria
Complex I
Complex III
IMS
Peroxidized
or damaged cardiolipin
Elamipretide
Restore normal mitochondrial function
Mitochondrial inner membrane
Complex IV
matrix

Retinal mitochondrial dysfunction associated with AMD pathobiology (human histology; mouse models)

Phase 1 study of elamipretide for dry AMD (ReCLAIM study) recently completed

Mitochondrial Dysfunction in Neovascular AMD?

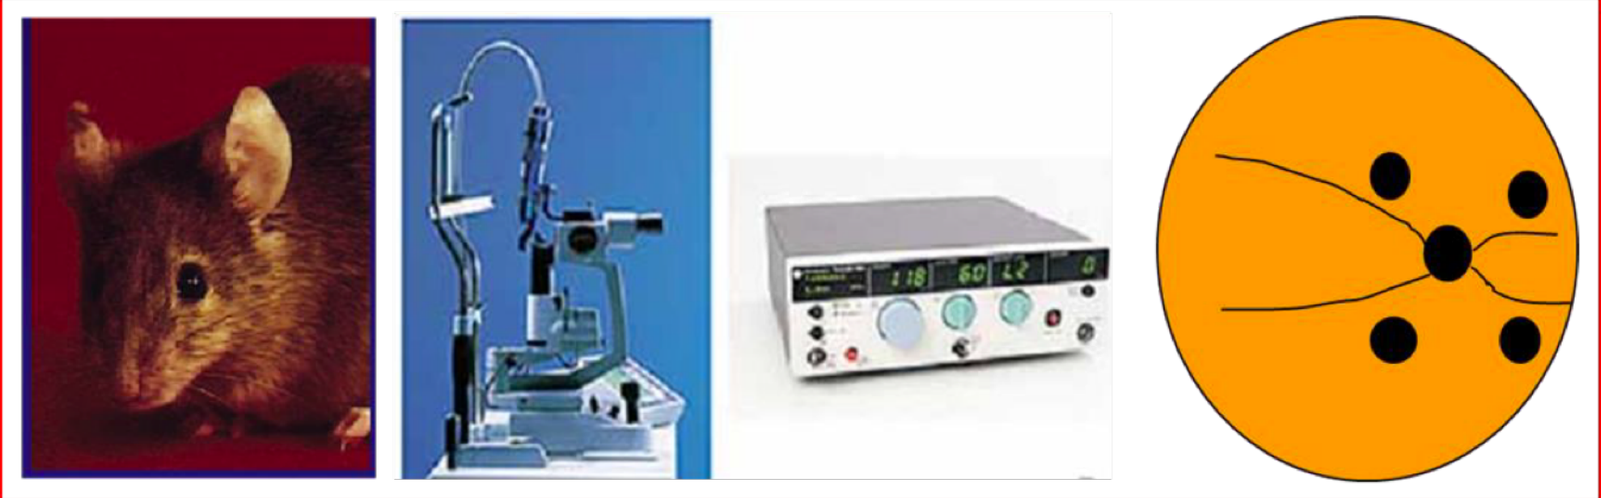

C57BL/6J mouse, 810 nm laser CNV model

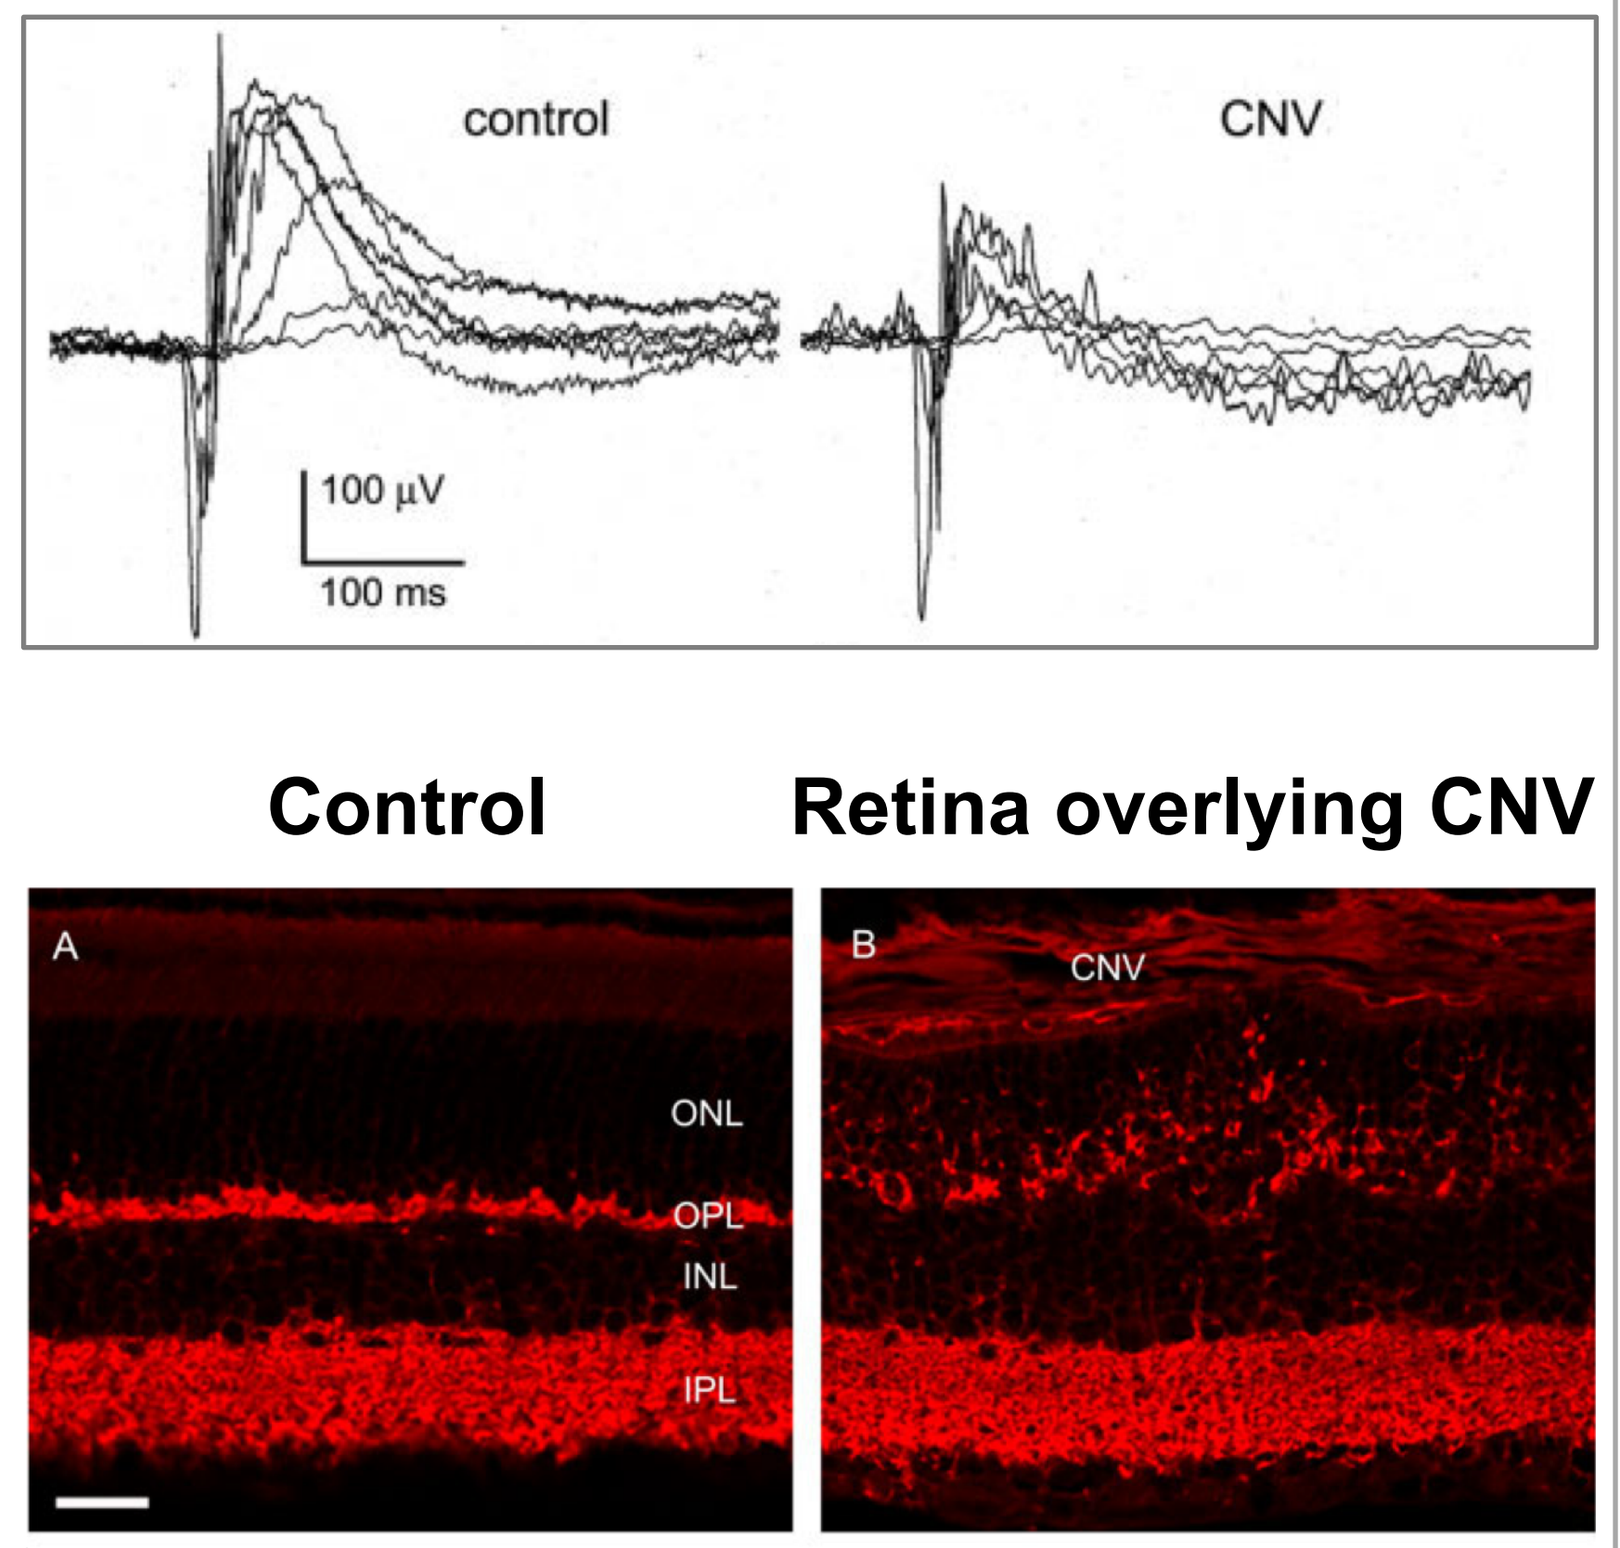
control
CNV
100 µV
100 ms
Control
Retina overlying CNV
A
B
ONL
OPL
INL
IPL

Fig. 1. Induction of laser CNV in 9 m.o. mice results in diminished ERG B-wave amplitudes (upper panel), and disrupted photoreceptor-bipolar synapses at OPL (SV2 staining, red, lower panel)

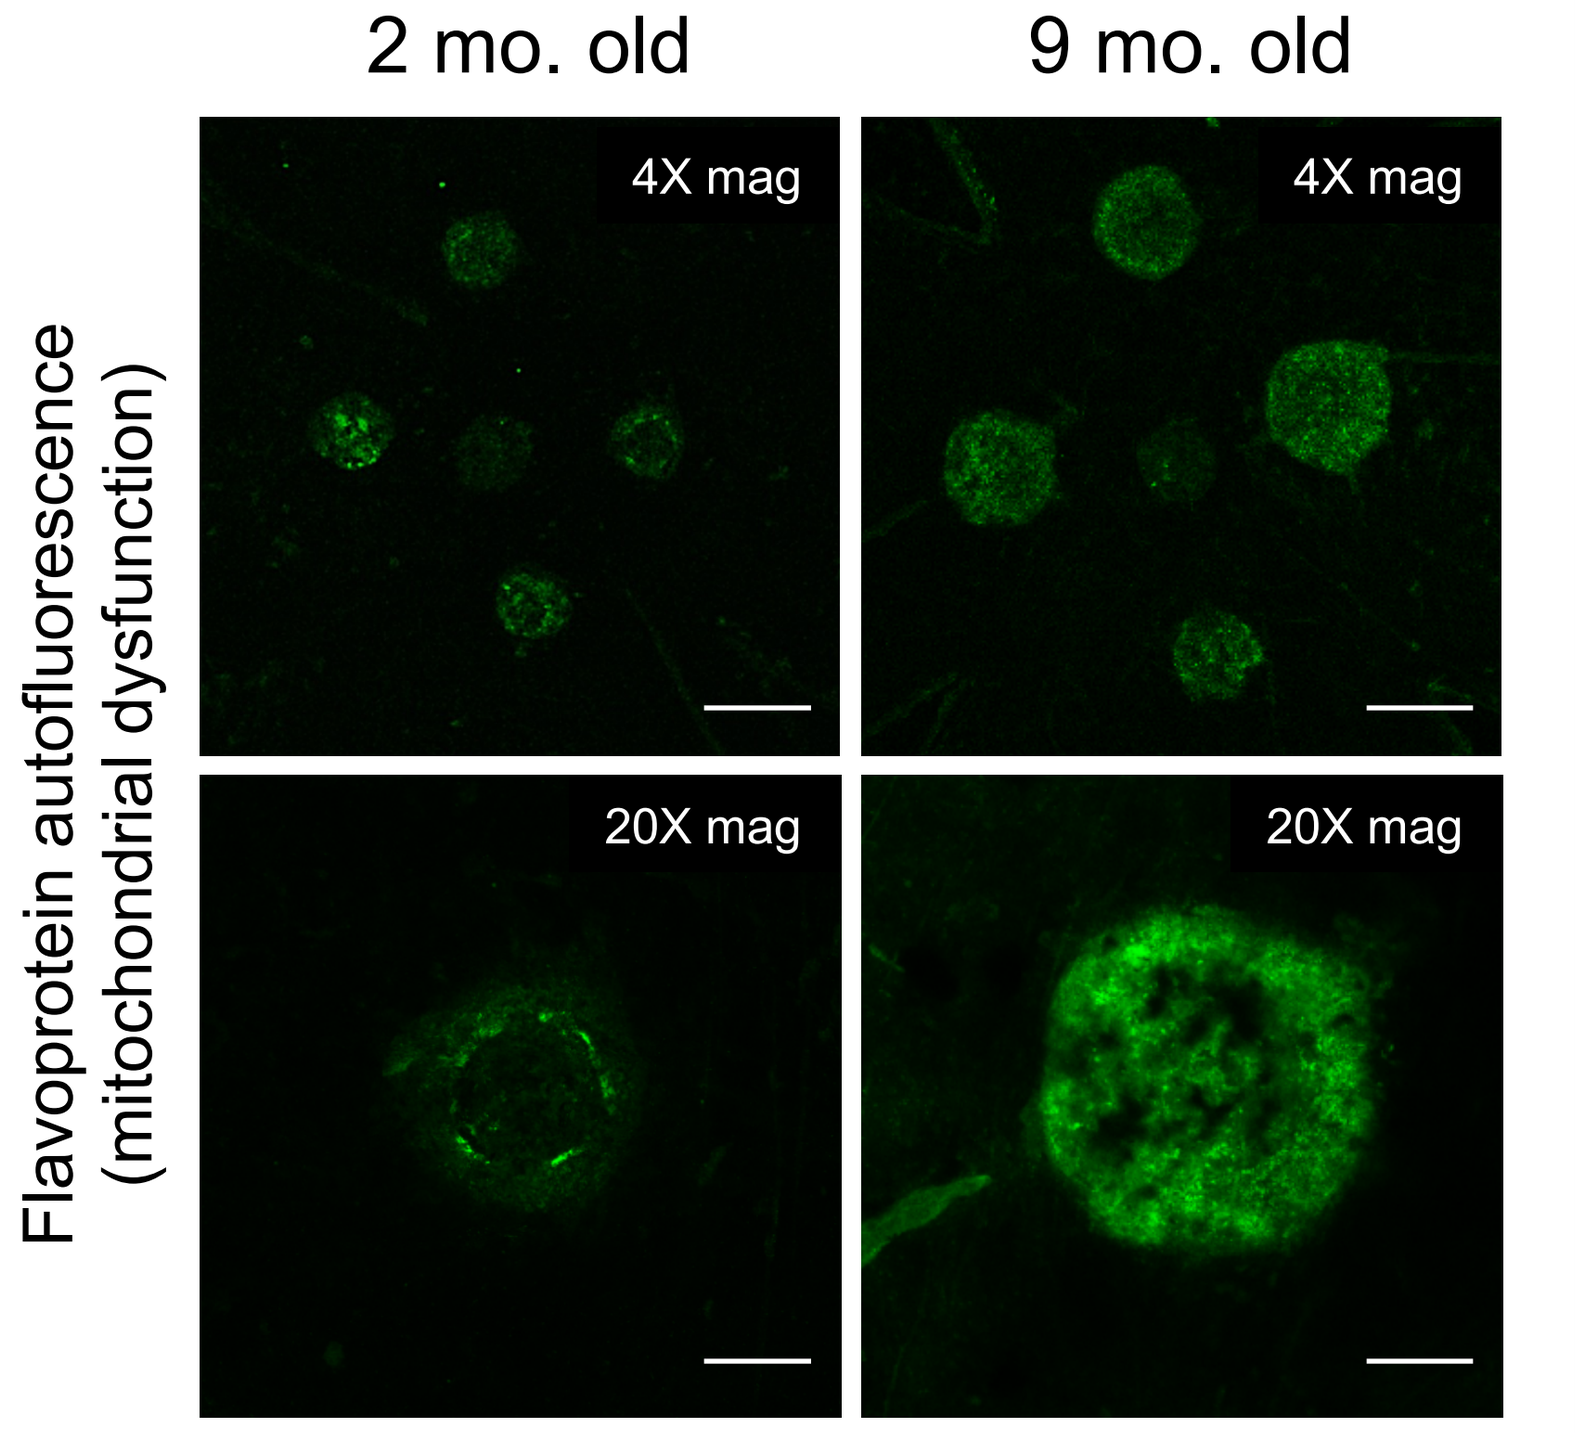
2 mo. old
9 mo. old
4X mag
20X mag
Flavoprotein autofluorescence (mitochondrial dysfunction)

Fig. 2. CNV in 9 m.o. mice - significant flavoprotein auto-fluorescence signal (mitochondrial dysfunction)

STUDY OBJECTIVE

Assess effects of systemic elamipretide on fellow, non-study eyes with quiescent NV AMD in ReCLAIM study, Ph. 1 study of elamipretide for dry AMD

DESIGN

- Phase 1, open-label, 24-week clinical trial (NCT02848313, Duke IRB approved) of subcutaneous elamipretide (40 mg) for dry AMD (drusen or noncentral GA). Assessments performed on both eyes.
- 13 **fellow eyes** with NV AMD; quiescent disease on anti-VEGF *or* stable inactive.
- Scar, untreated / persistent disease, new conversion, ocular surgery: not included.

Population Demographics (n=13)	
Mean Age	77.7 (66-95)
Percentage Female	46.2%
History of Smoking (Not Active)	53.8%
Mean No. of Anti-VEGF Injections	12.5 (5-44)

Question 1: Effects of Elamipretide on Standard Luminance Best-Corrected Visual Acuity (BCVA)?

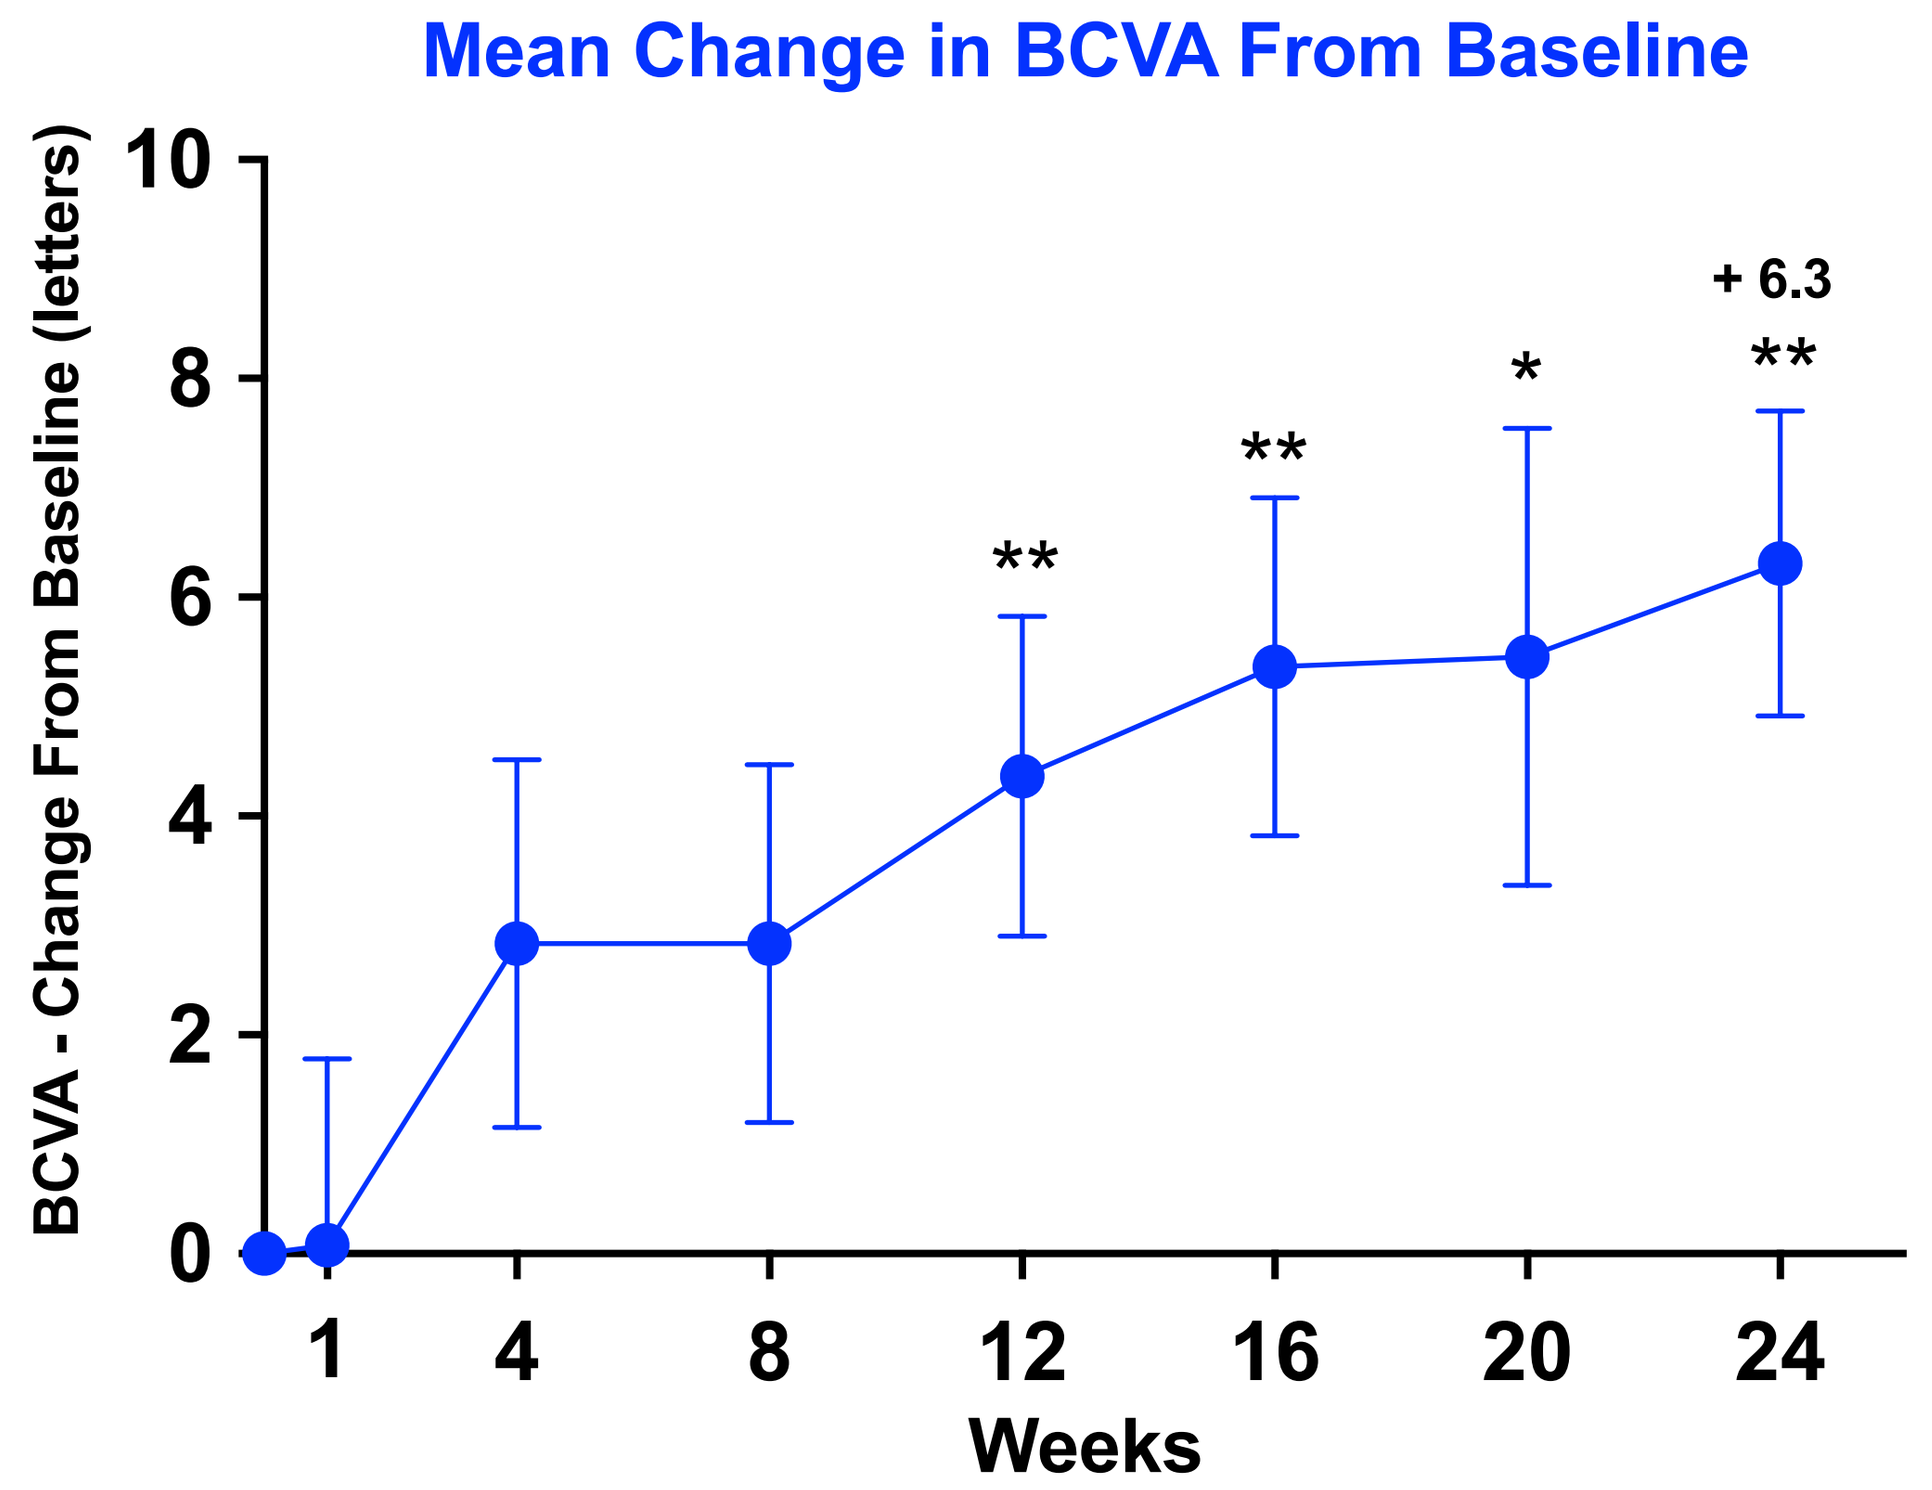
Mean Change in BCVA From Baseline
BCVA - Change From Baseline (letters)
Weeks
+ 6.3

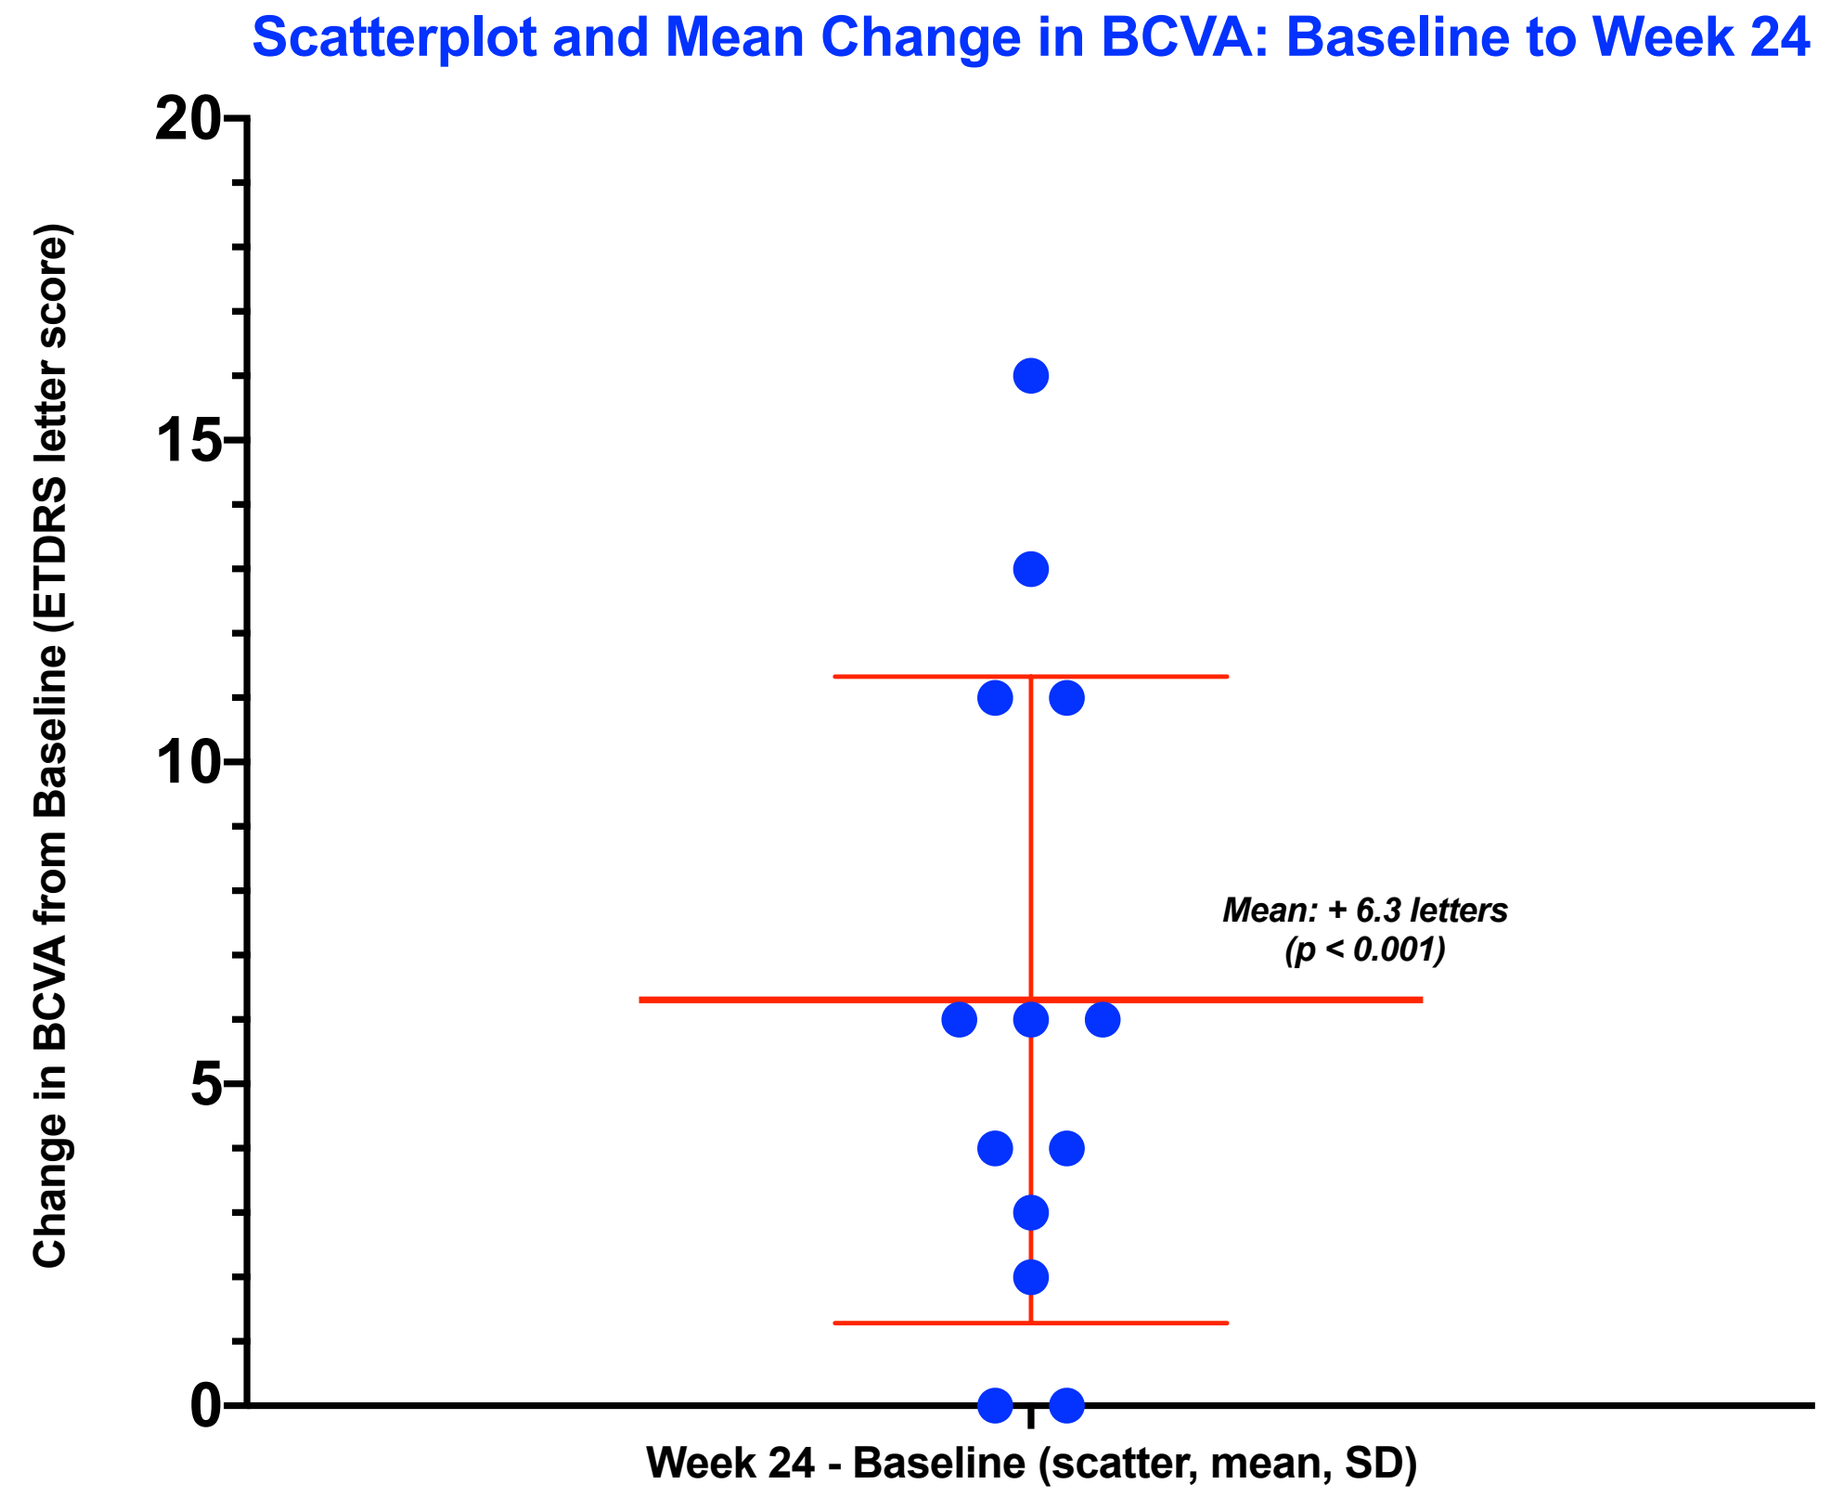
Scatterplot and Mean Change in BCVA: Baseline to Week 24
Change in BCVA from Baseline (ETDRS letter score)
Mean: + 6.3 letters
(p < 0.001)
Week 24 - Baseline (scatter, mean, SD)

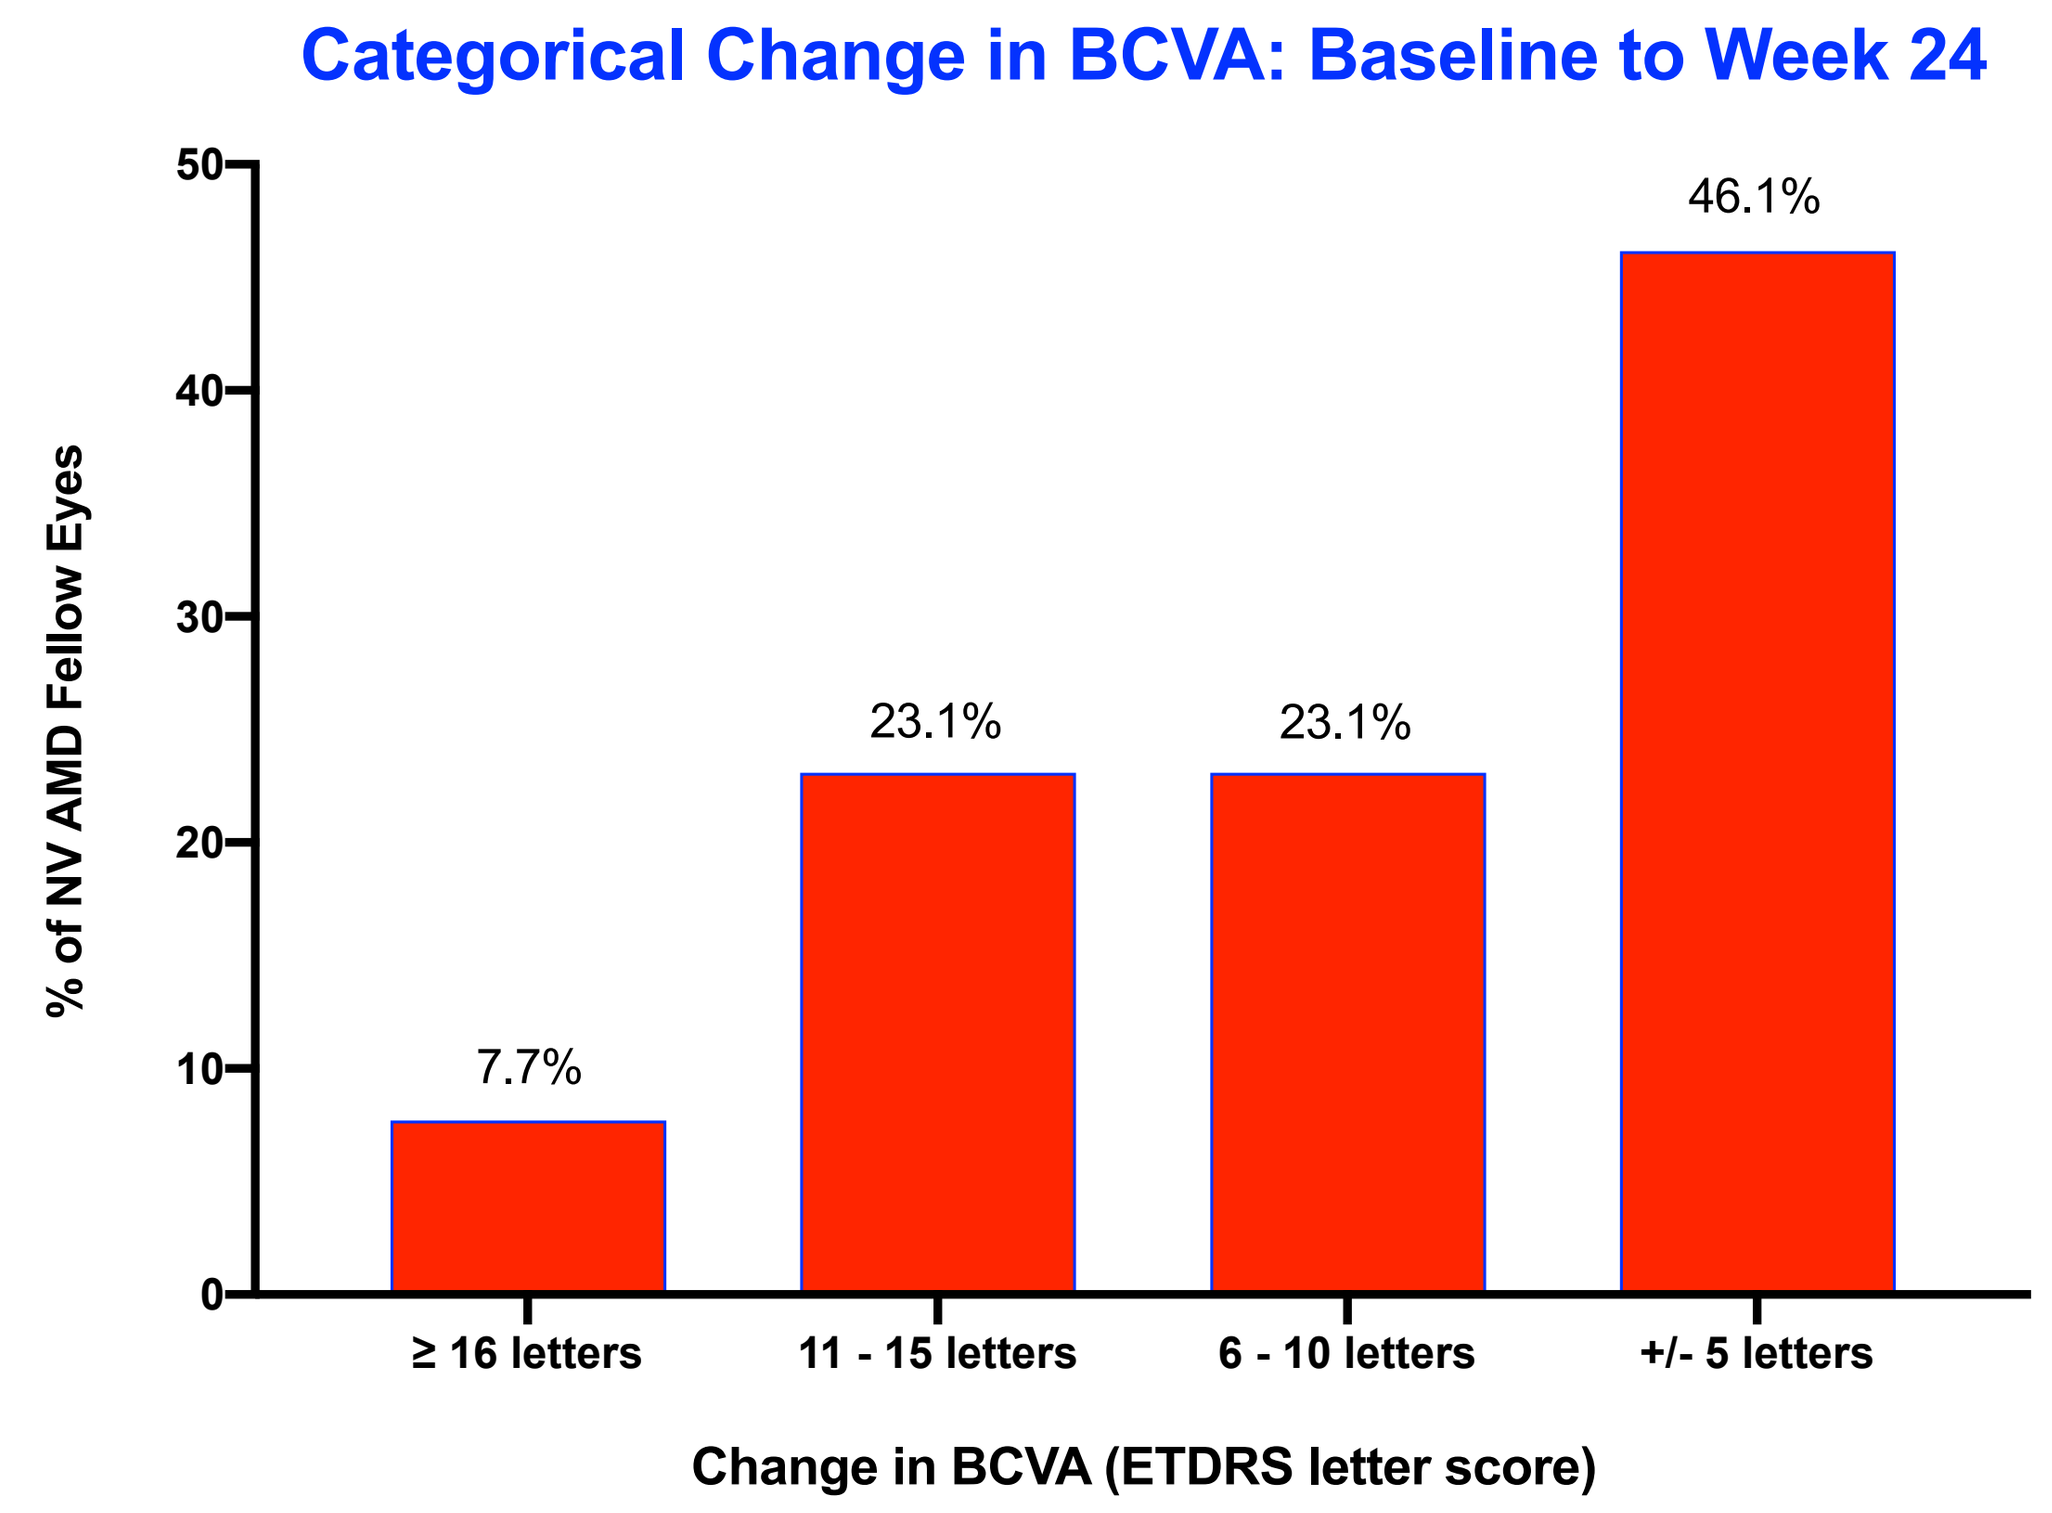
Categorical Change in BCVA: Baseline to Week 24
% of NV AMD Fellow Eyes
7.7%
23.1%
23.1%
46.1%
≥ 16 letters
11 - 15 letters
6 - 10 letters
+/- 5 letters
Change in BCVA (ETDRS letter score)

Question 2: Effects of Elamipretide on Low Luminance Best-Corrected Visual Acuity (LLVA)?

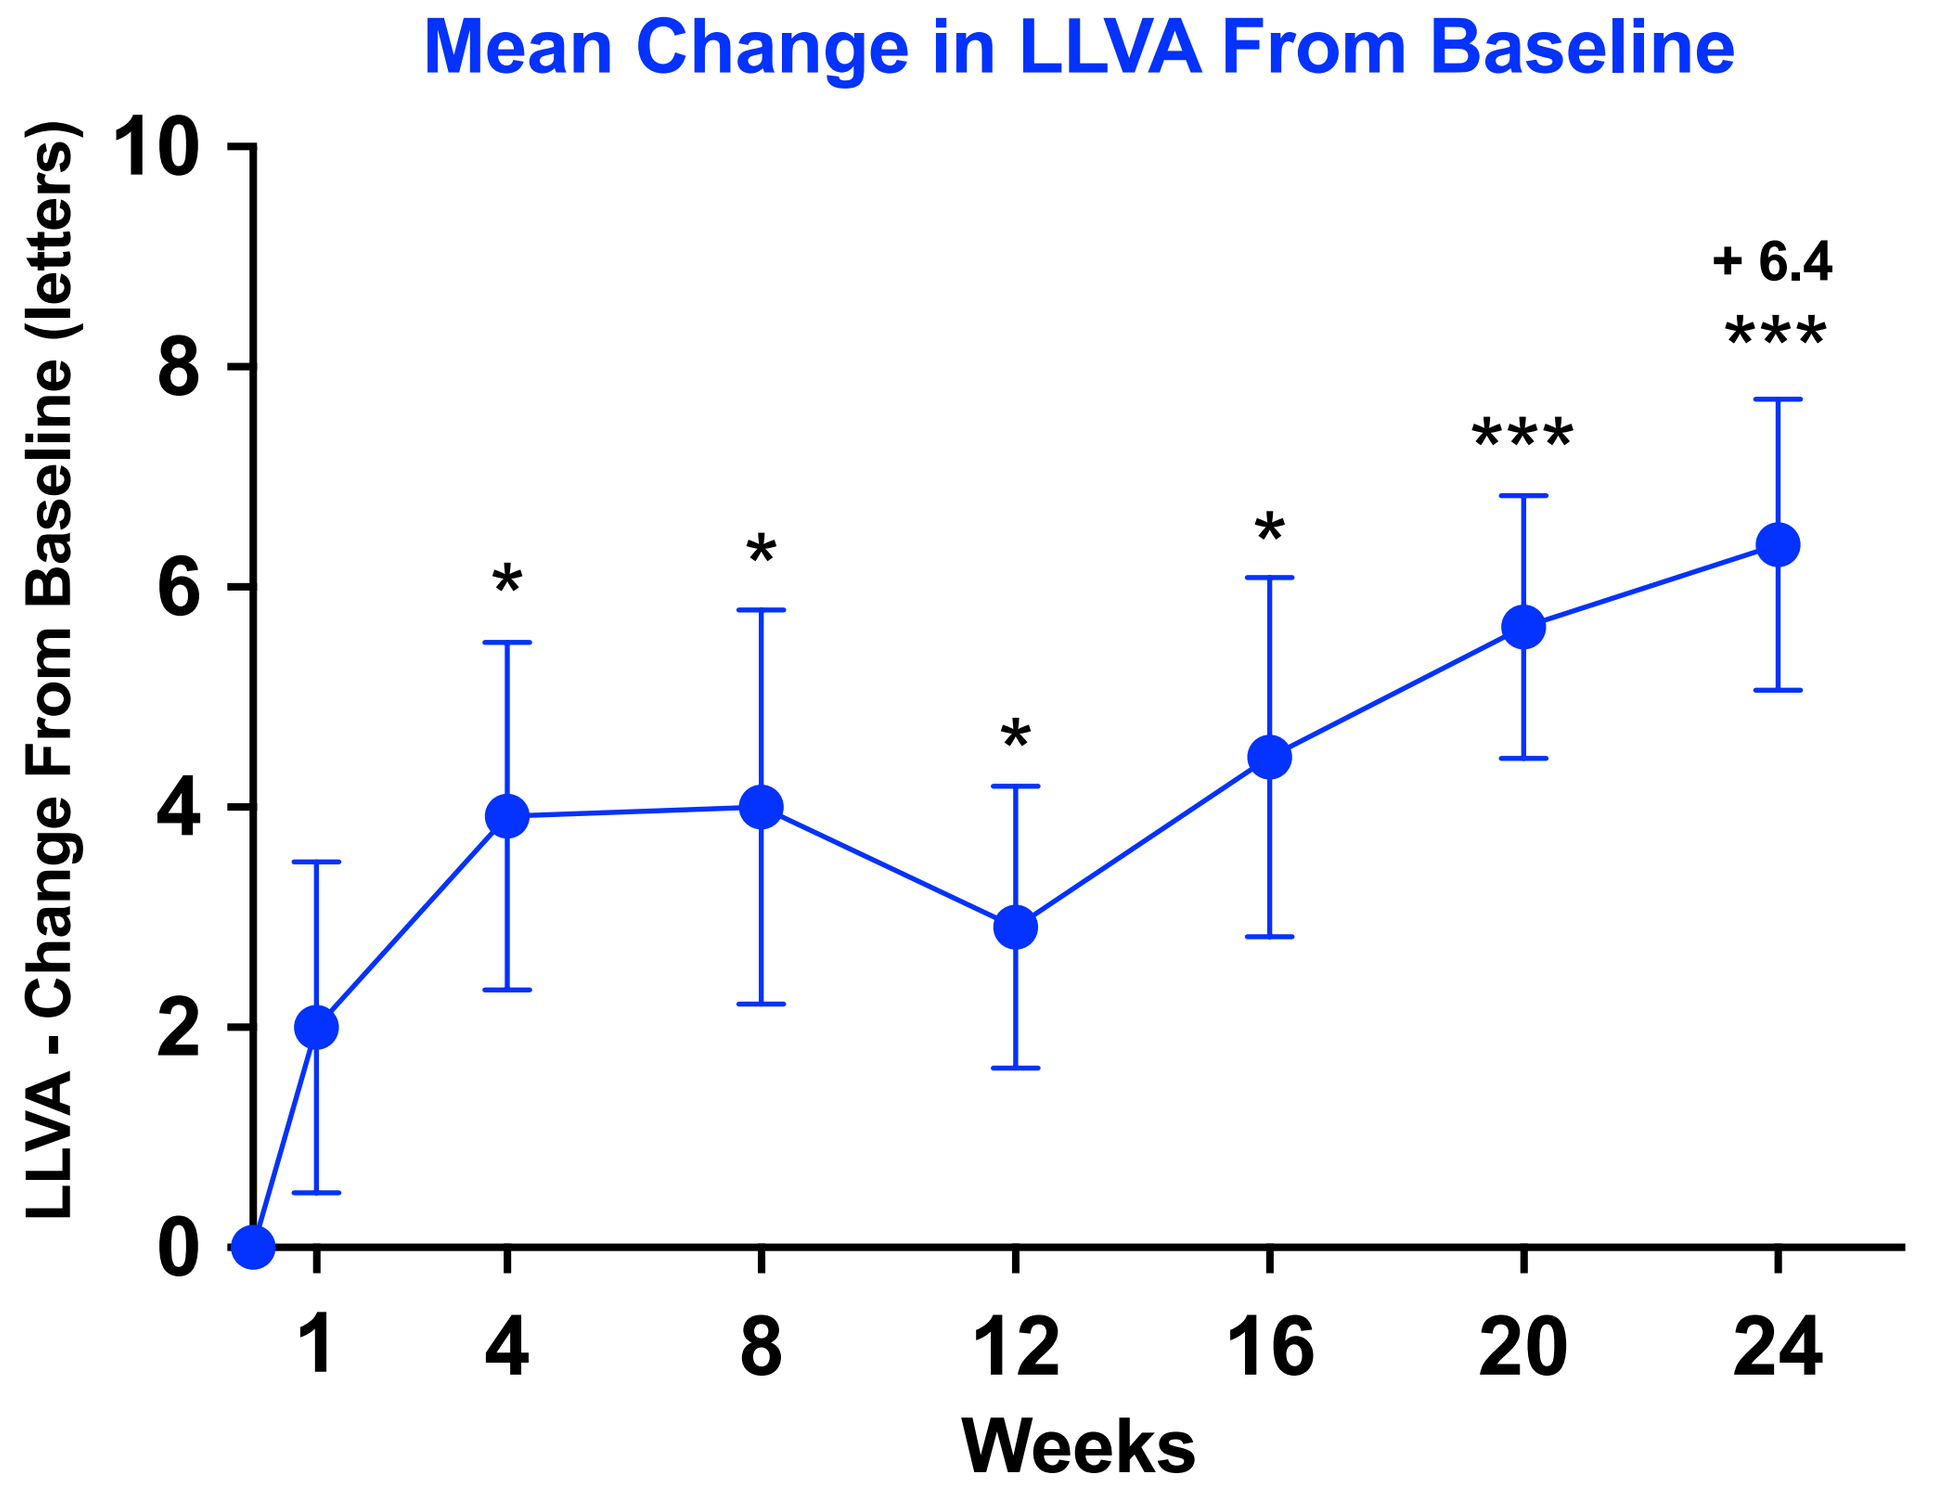
Mean Change in LLVA From Baseline
LLVA - Change From Baseline (letters)
Weeks
+ 6.4

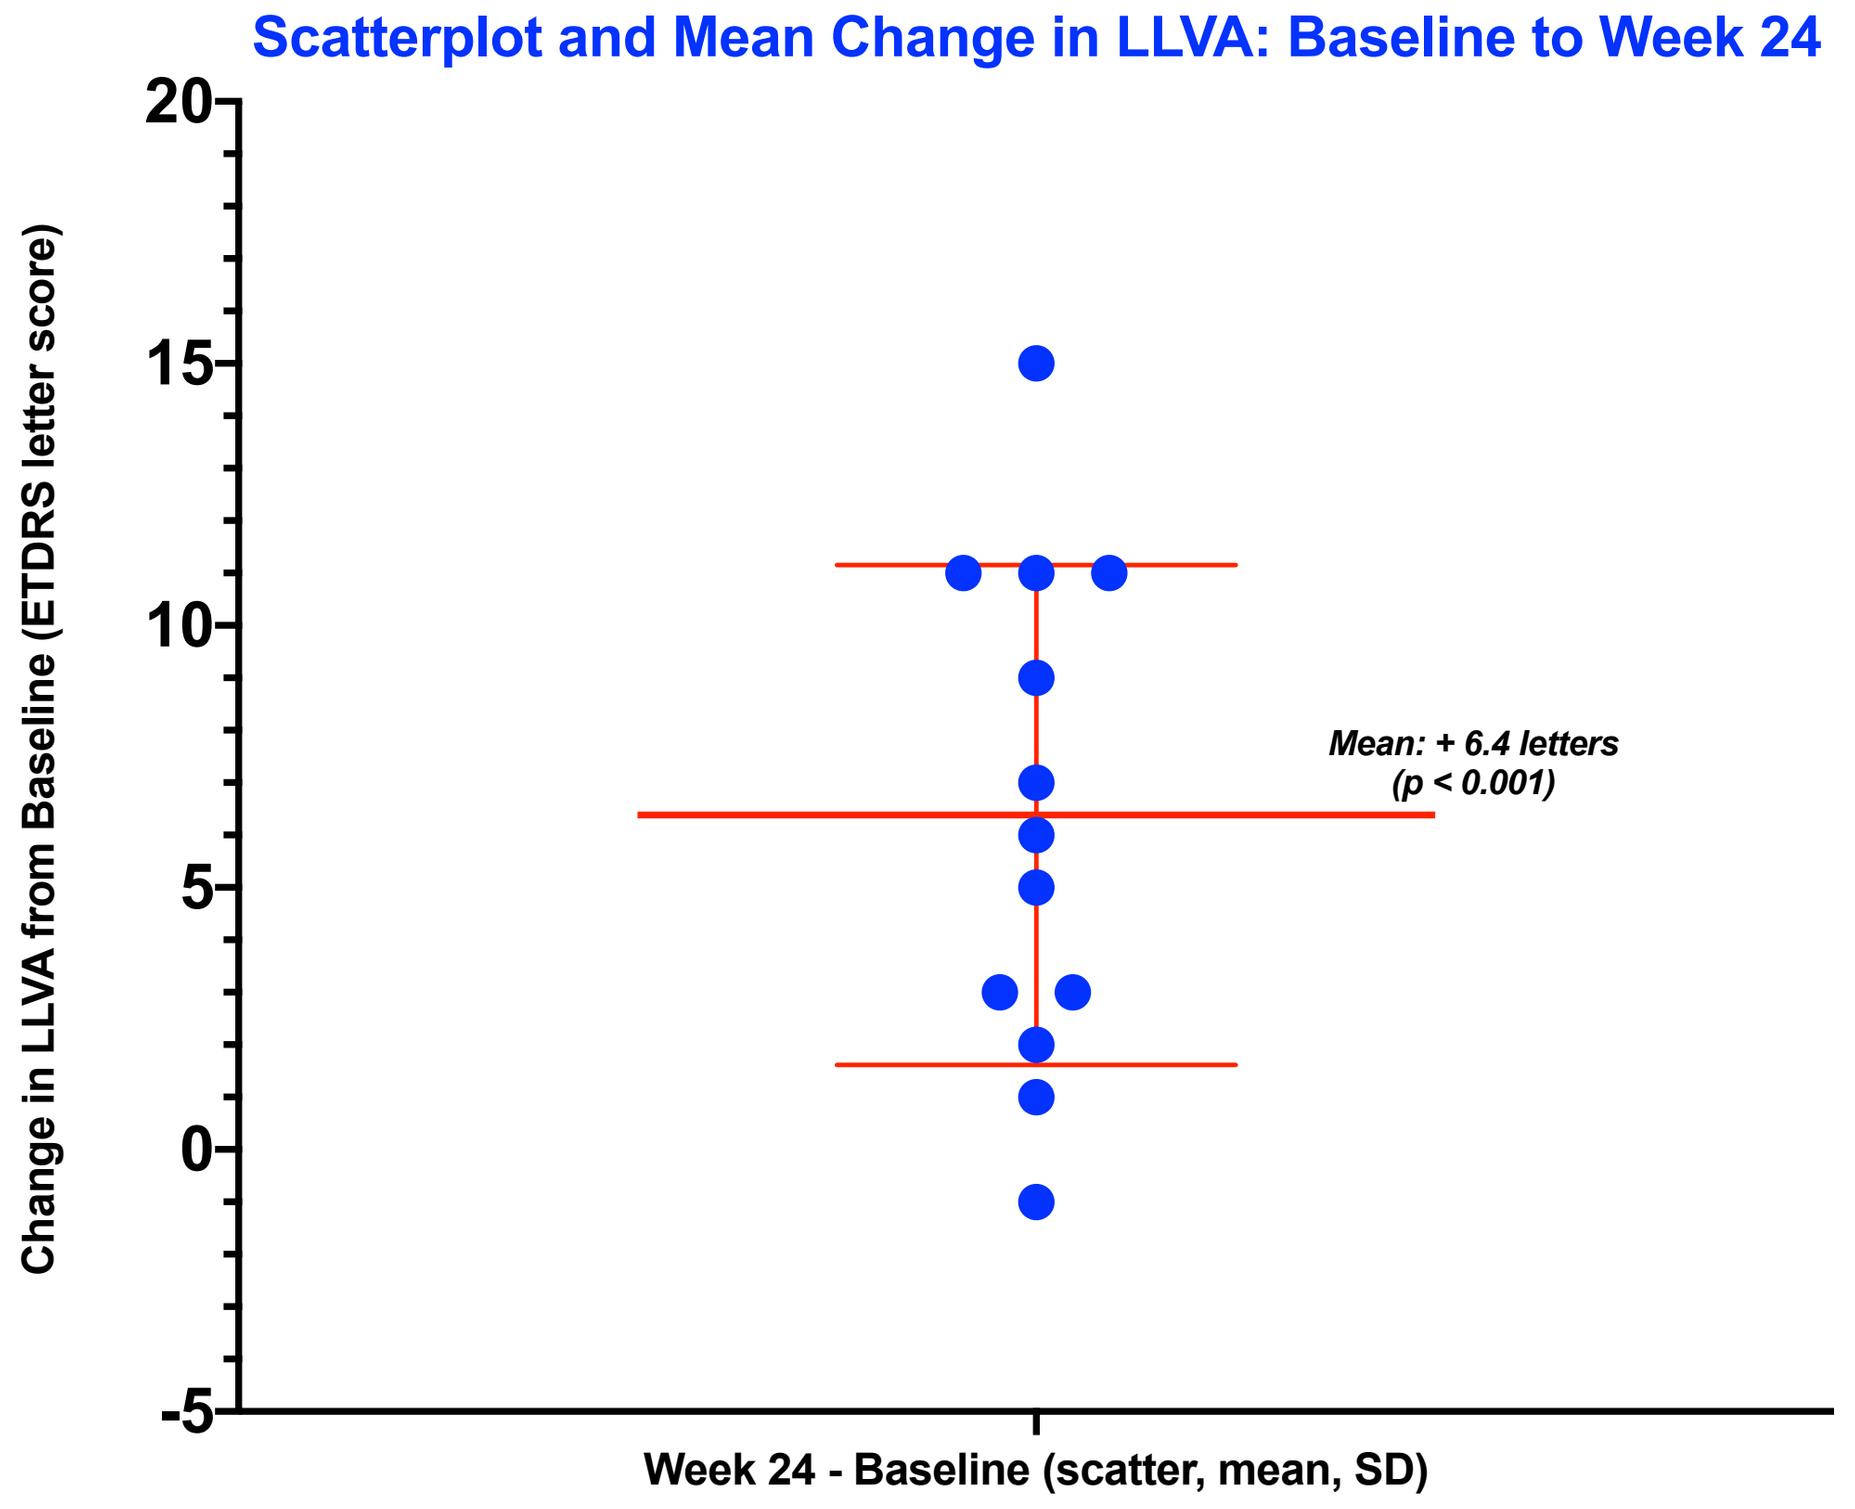
Scatterplot and Mean Change in LLVA: Baseline to Week 24
Change in LLVA from Baseline (ETDRS letter score)
Mean: + 6.4 letters
(p < 0.001)
Week 24 - Baseline (scatter, mean, SD)

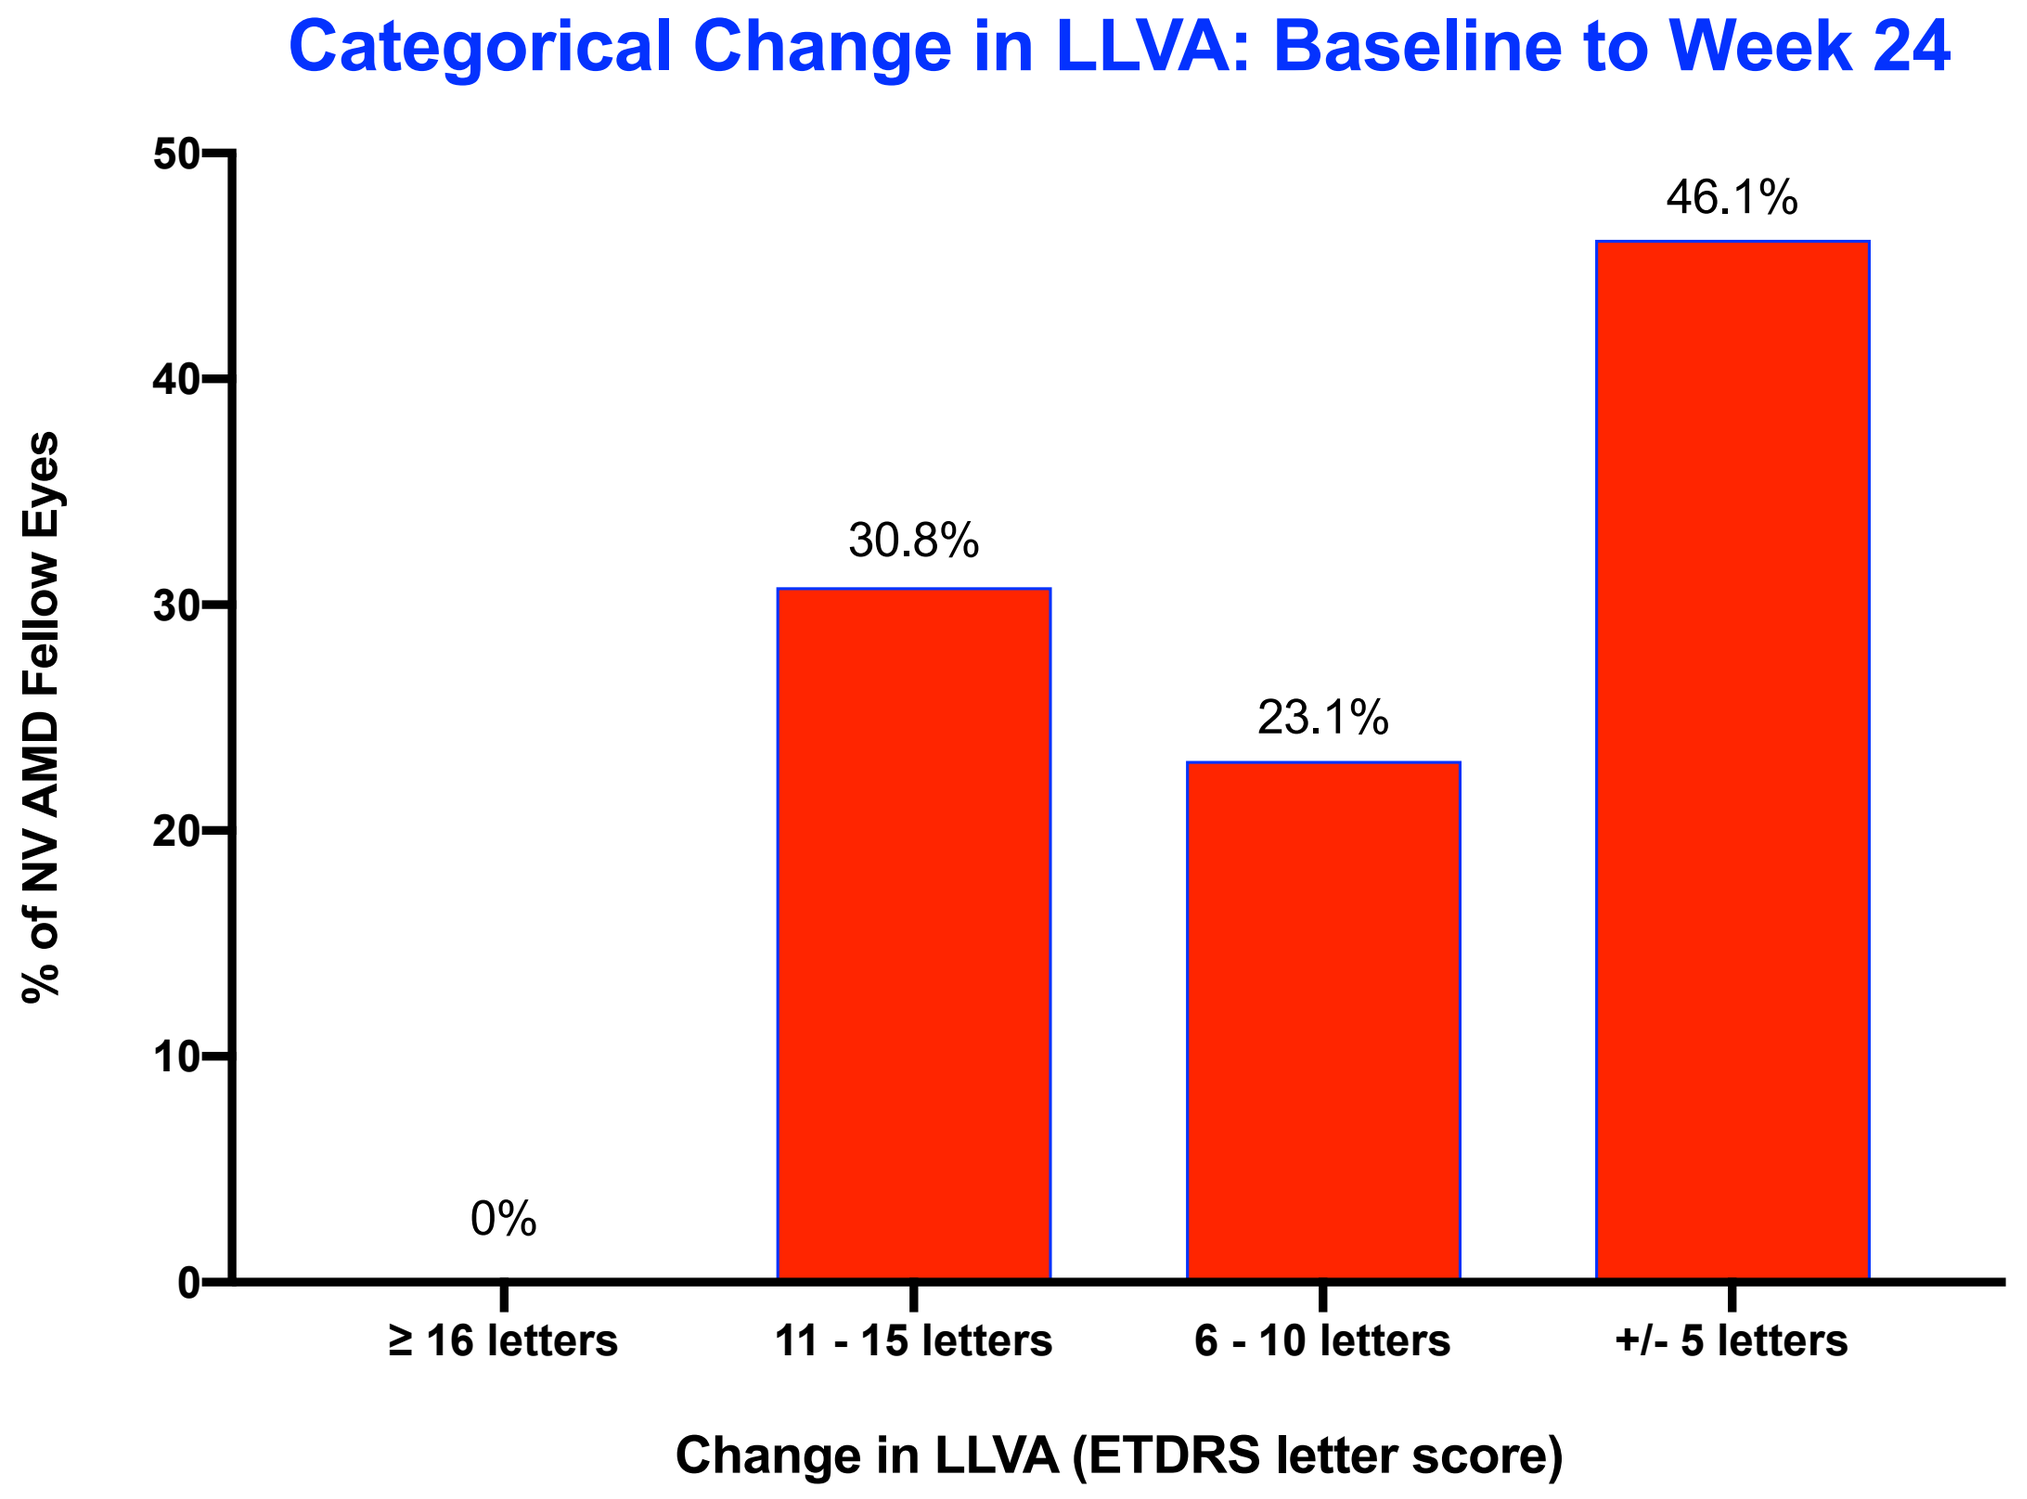
Categorical Change in LLVA: Baseline to Week 24
% of NV AMD Fellow Eyes
0%
30.8%
23.1%
46.1%
≥ 16 letters
11 - 15 letters
6 - 10 letters
+/- 5 letters
Change in LLVA (ETDRS letter score)

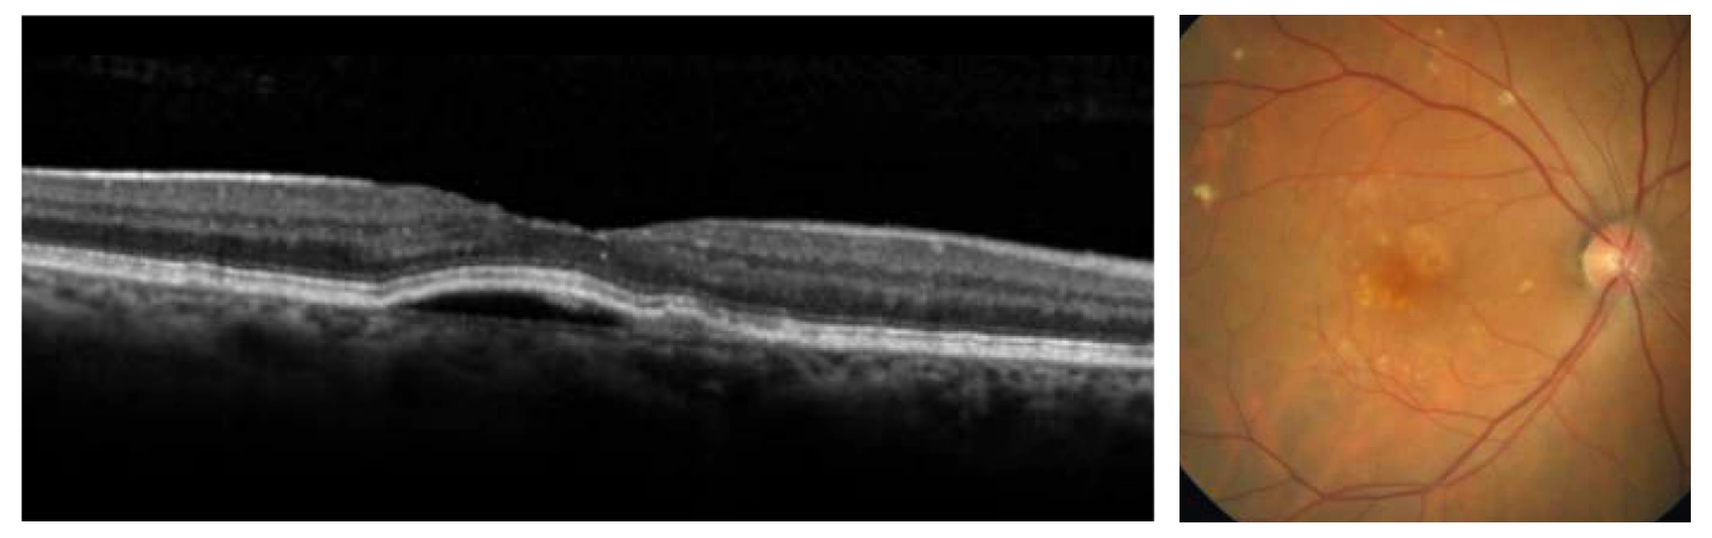

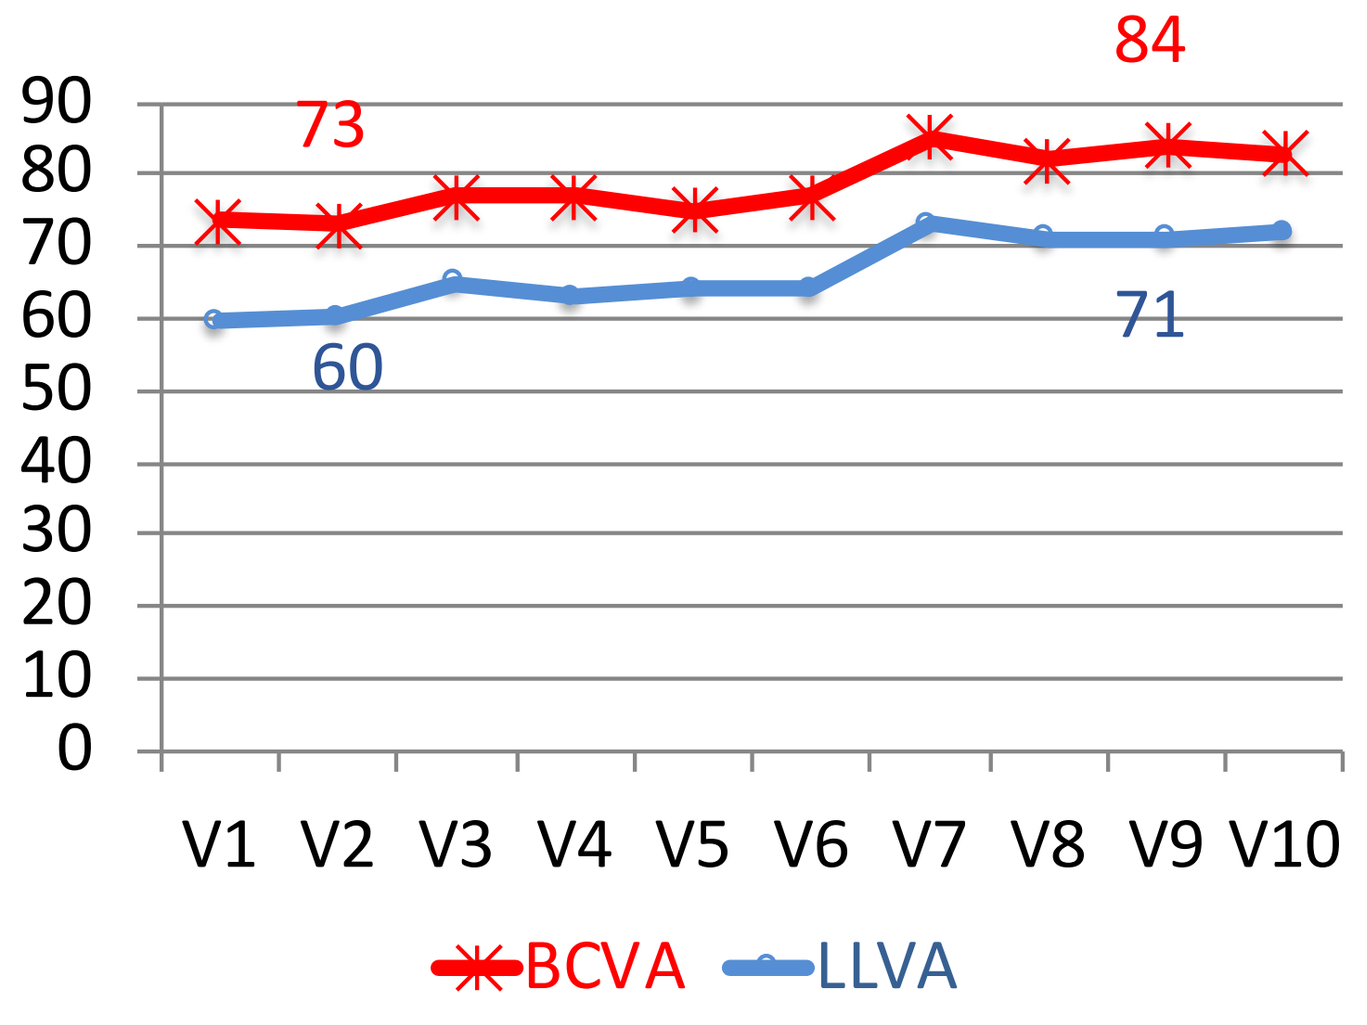
73
84
60
71
V1 V2 V3 V4 V5 V6 V7 V8 V9 V10
BCVA
LLVA

Fig. 3. Case example of fellow eye with NV AMD, which was stably quiescent on q8week ranibizumab during the study. Both BCVA and LLVA (ETDRS letter scores) improved while on elamipretide (right graph).

CONCLUSIONS

- Subcutaneous elamipretide, mitochondria-targeted drug, was associated with improved vision in fellow, non-study eyes with NV AMD in the Phase 1 ReCLAIM study for dry AMD
- Further study is warranted to assess efficacy of elamipretide as novel therapy for leakage-independent vision loss in NV AMD

This work was funded by a sponsored research agreement from Stealth BioTherapeutics to Duke University. PSM, MJA, and SWC consultants to Stealth.