Elamipretide in Patients with Barth Syndrome: A Randomized, Double-Blind, Placebo-Controlled Clinical Trial Followed by 36-Week Open-Label Extension

William Reid Thompson[1], Ryan Manuel[2], Anthony Aiudi[3], John J. Jones[3], Jim Carr[3], Brittany Hornby[4], Hilary J. Vernon[2]

[1]Department of Pediatric Cardiology, Taussig Heart Center, Johns Hopkins University School of Medicine, Baltimore, Maryland, [2]Department of Genetic Medicine, Johns Hopkins University School of Medicine, Baltimore, MD; [3]Stealth BioTherapeutics, Inc, Newton, MA; [4]Department of Physical Therapy, Kennedy Krieger, Baltimore, MD

INTRODUCTION

- Barth syndrome (BTHS) is a rare, X-linked disease caused by defects in TAZ, the tafazzin encoding gene, responsible for the final remodeling step to mature cardiolipin, critical for mitochondrial function
- The inability to produce mature cardiolipin leads to clinical manifestations of BTHS, including cardiac and skeletal myopathy, neutropenia, and growth abnormalities
- Elamipretide localizes to the inner mitochondrial membrane, where it is believed to associate with cardiolipin, improving membrane stability and ATP production and reducing pathogenic ROS production
- The efficacy and safety of elamipretide are being studied in TAZPOWER, which is the first clinical trial to evaluate a therapeutic agent in patients with BTHS

OBJECTIVE

- To measure efficacy through functional, patient-reported outcome (PRO) assessments, and cardiac parameters and safety/tolerability through adverse events (AEs) and laboratory tests

METHODS

Study Design

- TAZPOWER is a 28-week, randomized, double-blind, placebo-controlled trial followed by an open-label extension (OLE)

Figure 1. Study Design

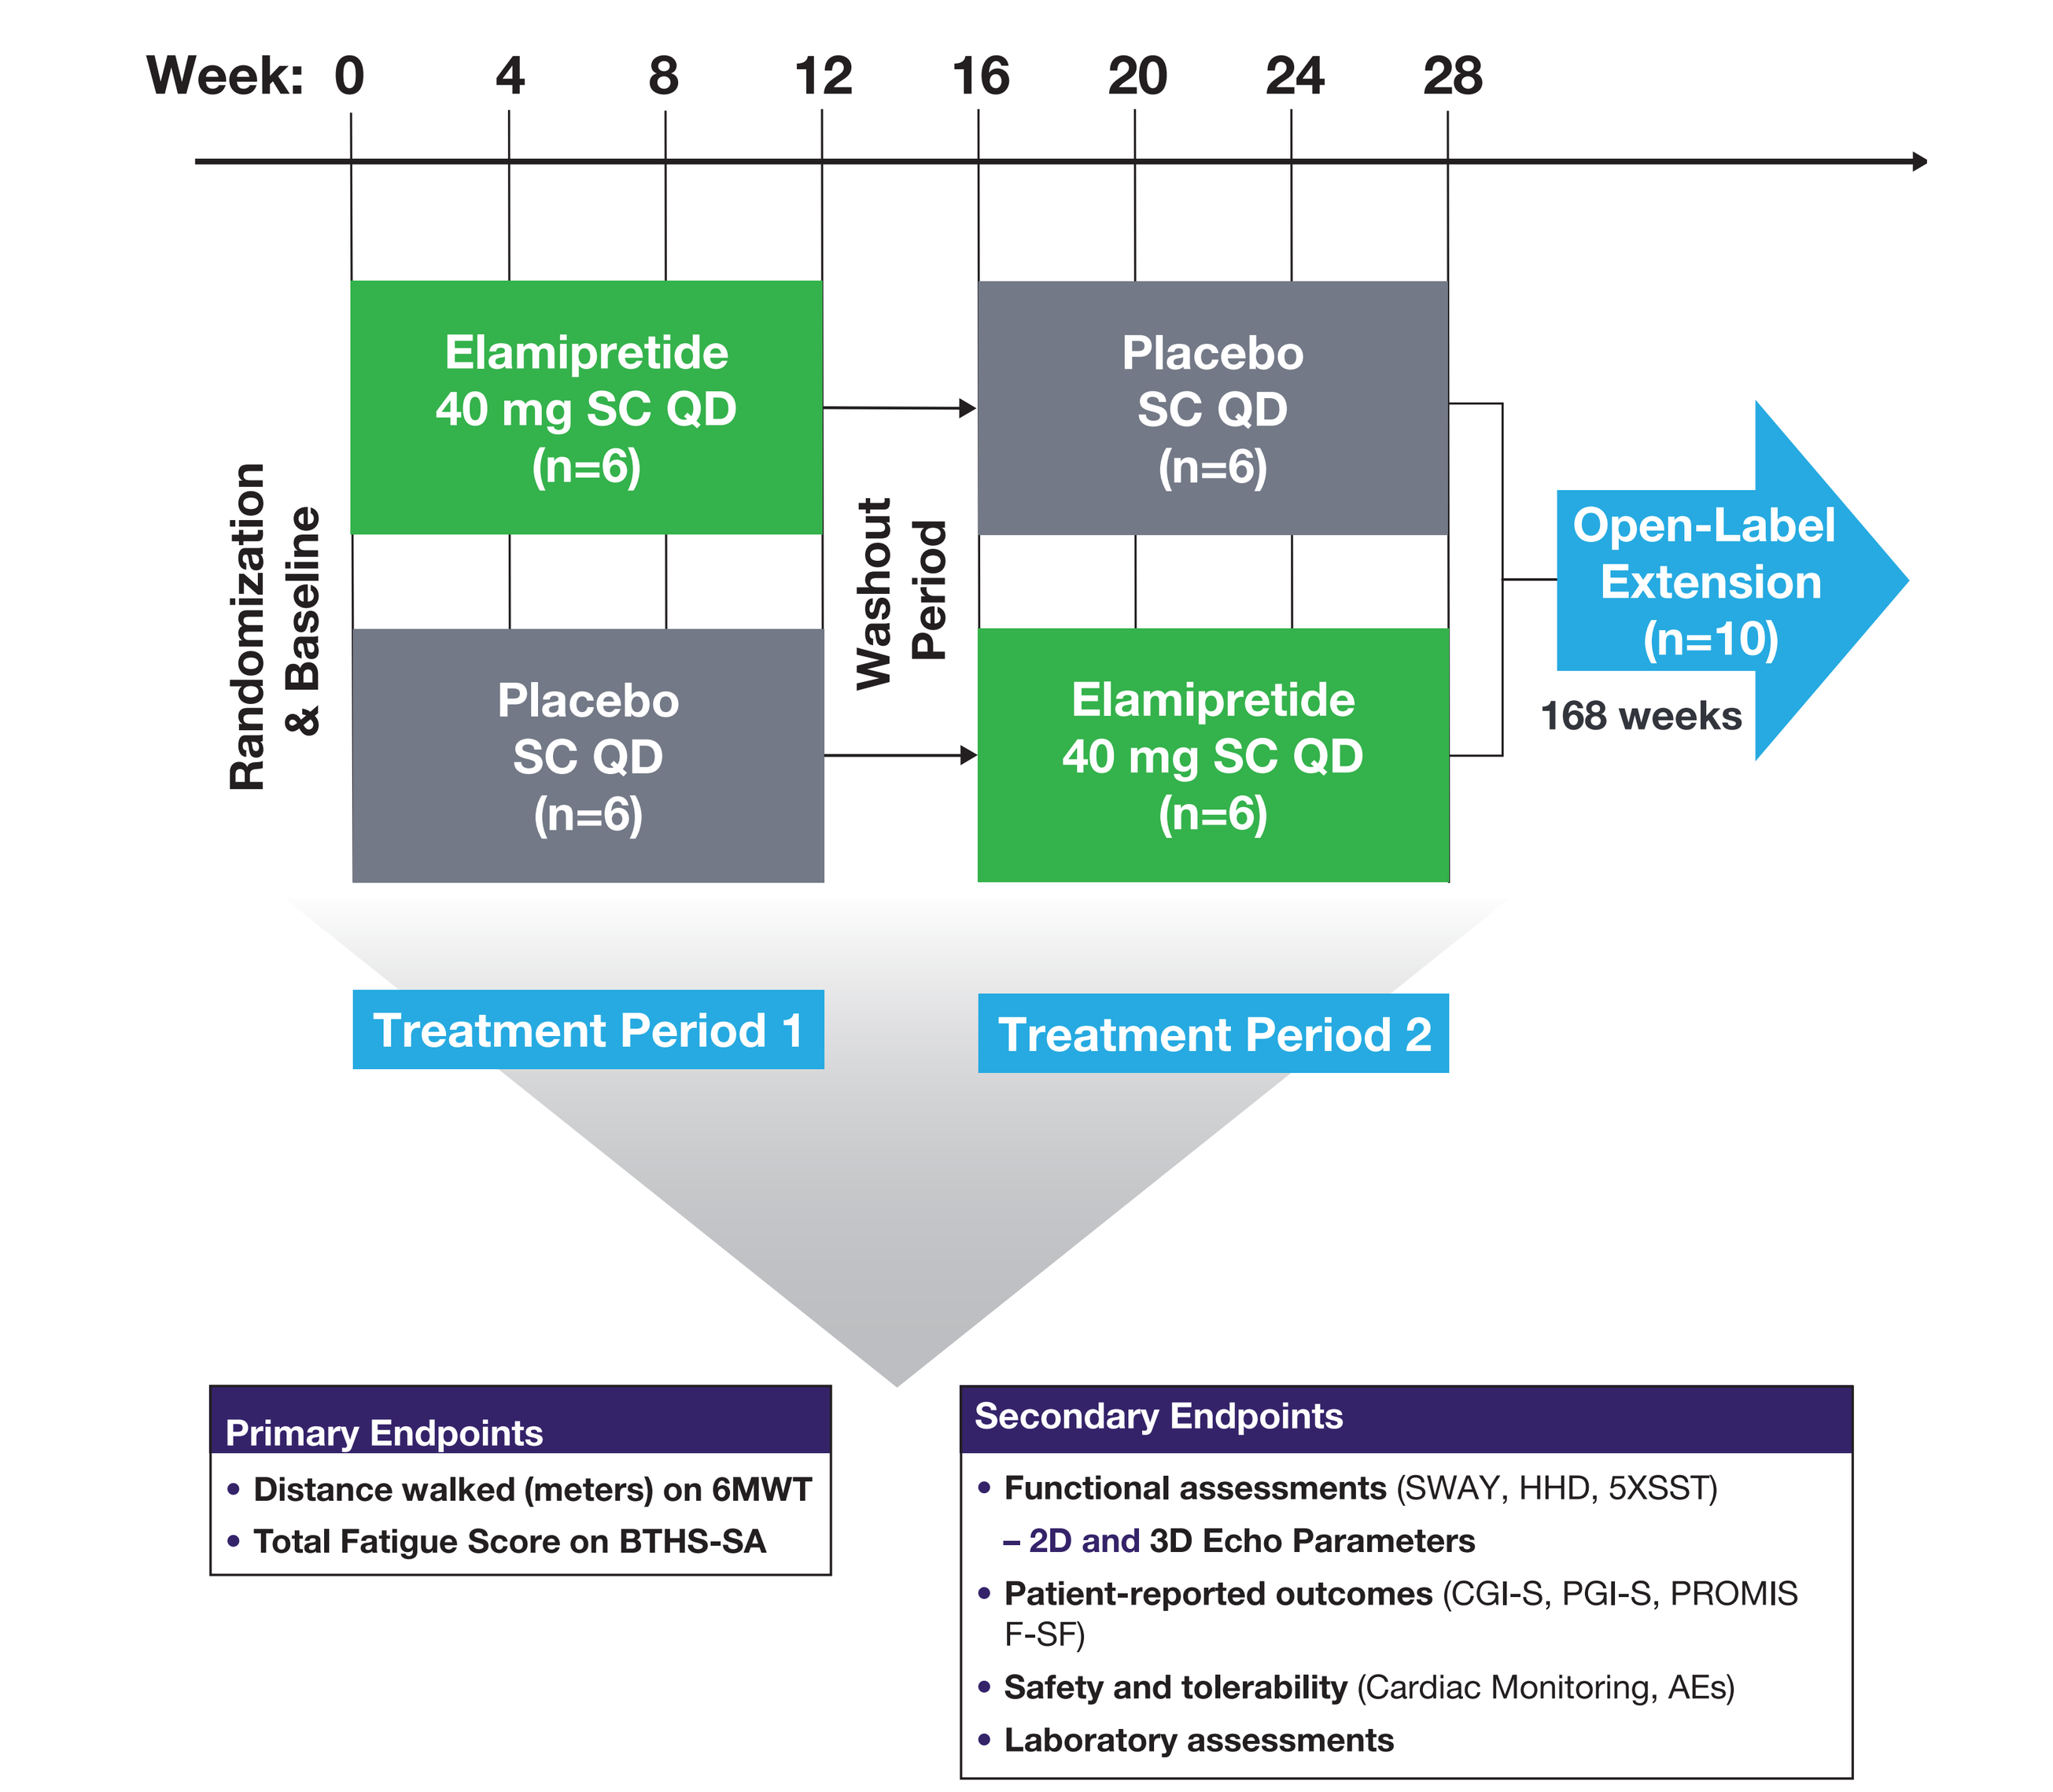
Week: 0 4 8 12 16 20 24 28
Randomization & Baseline
Elamipretide 40 mg SC QD (n=6)
Placebo SC QD (n=6)
Washout Period
Placebo SC QD (n=6)
Elamipretide 40 mg SC QD (n=6)
Open-Label Extension (n=10)
168 weeks
Treatment Period 1
Treatment Period 2
Primary Endpoints
• Distance walked (meters) on 6MWT
• Total Fatigue Score on BTHS-SA
Secondary Endpoints
• Functional assessments (SWAY, HHD, 5XSST)
– 2D and 3D Echo Parameters
• Patient-reported outcomes (CGI-S, PGI-S, PROMIS F-SF)
• Safety and tolerability (Cardiac Monitoring, AEs)
• Laboratory assessments

AEs=Adverse Events; BTHS-SA=BarTH Syndrome Symptom Assessment; CGI-S=Clinician Global Impression of Symptom Severity and Change Scale; 5XSST=Five Times Sit-to-Stand Test; HHD=Hand held Dynamometry; PGI-S=Patient Global Impression of Symptom Severity and Change Scale; PROMIS F-SF=Patient-Reported Outcomes Measurement Information System Fatigue-Short Form; 6MWT=6-Minute Walk Test; SWAY=SWAY Application Balance Assessment.

Key Inclusion Criteria

- Patients ≥12 years of age were required to have genetically confirmed BTHS, to be ambulatory but impaired as assessed by the 6MWT, and on stable medications
 - Body weight >30 kg, eGFR ≥90 mL/min or body weight >40 kg, eGFR ≥60 mL/min at screening

Key Exclusion Criteria

- Patients were excluded if they had been hospitalized within 30 days, had uncontrolled hypertension, a history of heart transplantation, or implantation of a cardioverter defibrillator within 3 months or expected implantation during the study

RESULTS

Patient Demographics

- A total of 12 patients were randomized into the trial

Table 1. Patient Demographics (N=12)

Demographic Variable	Demographic Result
Mean Age (years), (Range)	19.5 (12-35)
Male (n)	12
Race (n) White Multiracial	 11 1
Ethnicity (n) Not Hispanic or Latino Hispanic or Latino	 12 0
Mean Height (cm) (range)	167.3 (150.4-187.7)
Mean Weight (kg) (range)	50.8 (31.4-85.9)
BMI (kg/m^2) (range)	17.6 (13.6-24.4)
Mean 6MWT (meters)	406.35
Mean BTHS-SA Total Fatigue	7.56
Mean 3D LV End-Diastolic Volumes (EDV) (Z-score [SD])	-2.0 (1.36)
Mean 3D LV End-Systolic Volume (ESV) (Z-score [SD])	-1.79 (0.99)
Mean 3D LV Stroke Volume (Z-score [SD])	-1.84 (1.53)
Mean Ejection Fraction (% [SD])	60.6 (4.0)

Efficacy and Patient-Reported Outcomes

- At the end of the double-blind phase of the TAZPOWER trial, statistical significance was not achieved in the ITT population on the primary endpoints
- A total of 10 patients elected to continue into the TAZPOWER open-label extension (OLE) portion of the trial, which is currently ongoing (36 week OLE data currently available for 8 patients)

Figure 2. Functional and Patient-Reported Outcomes Assessments (n=8)

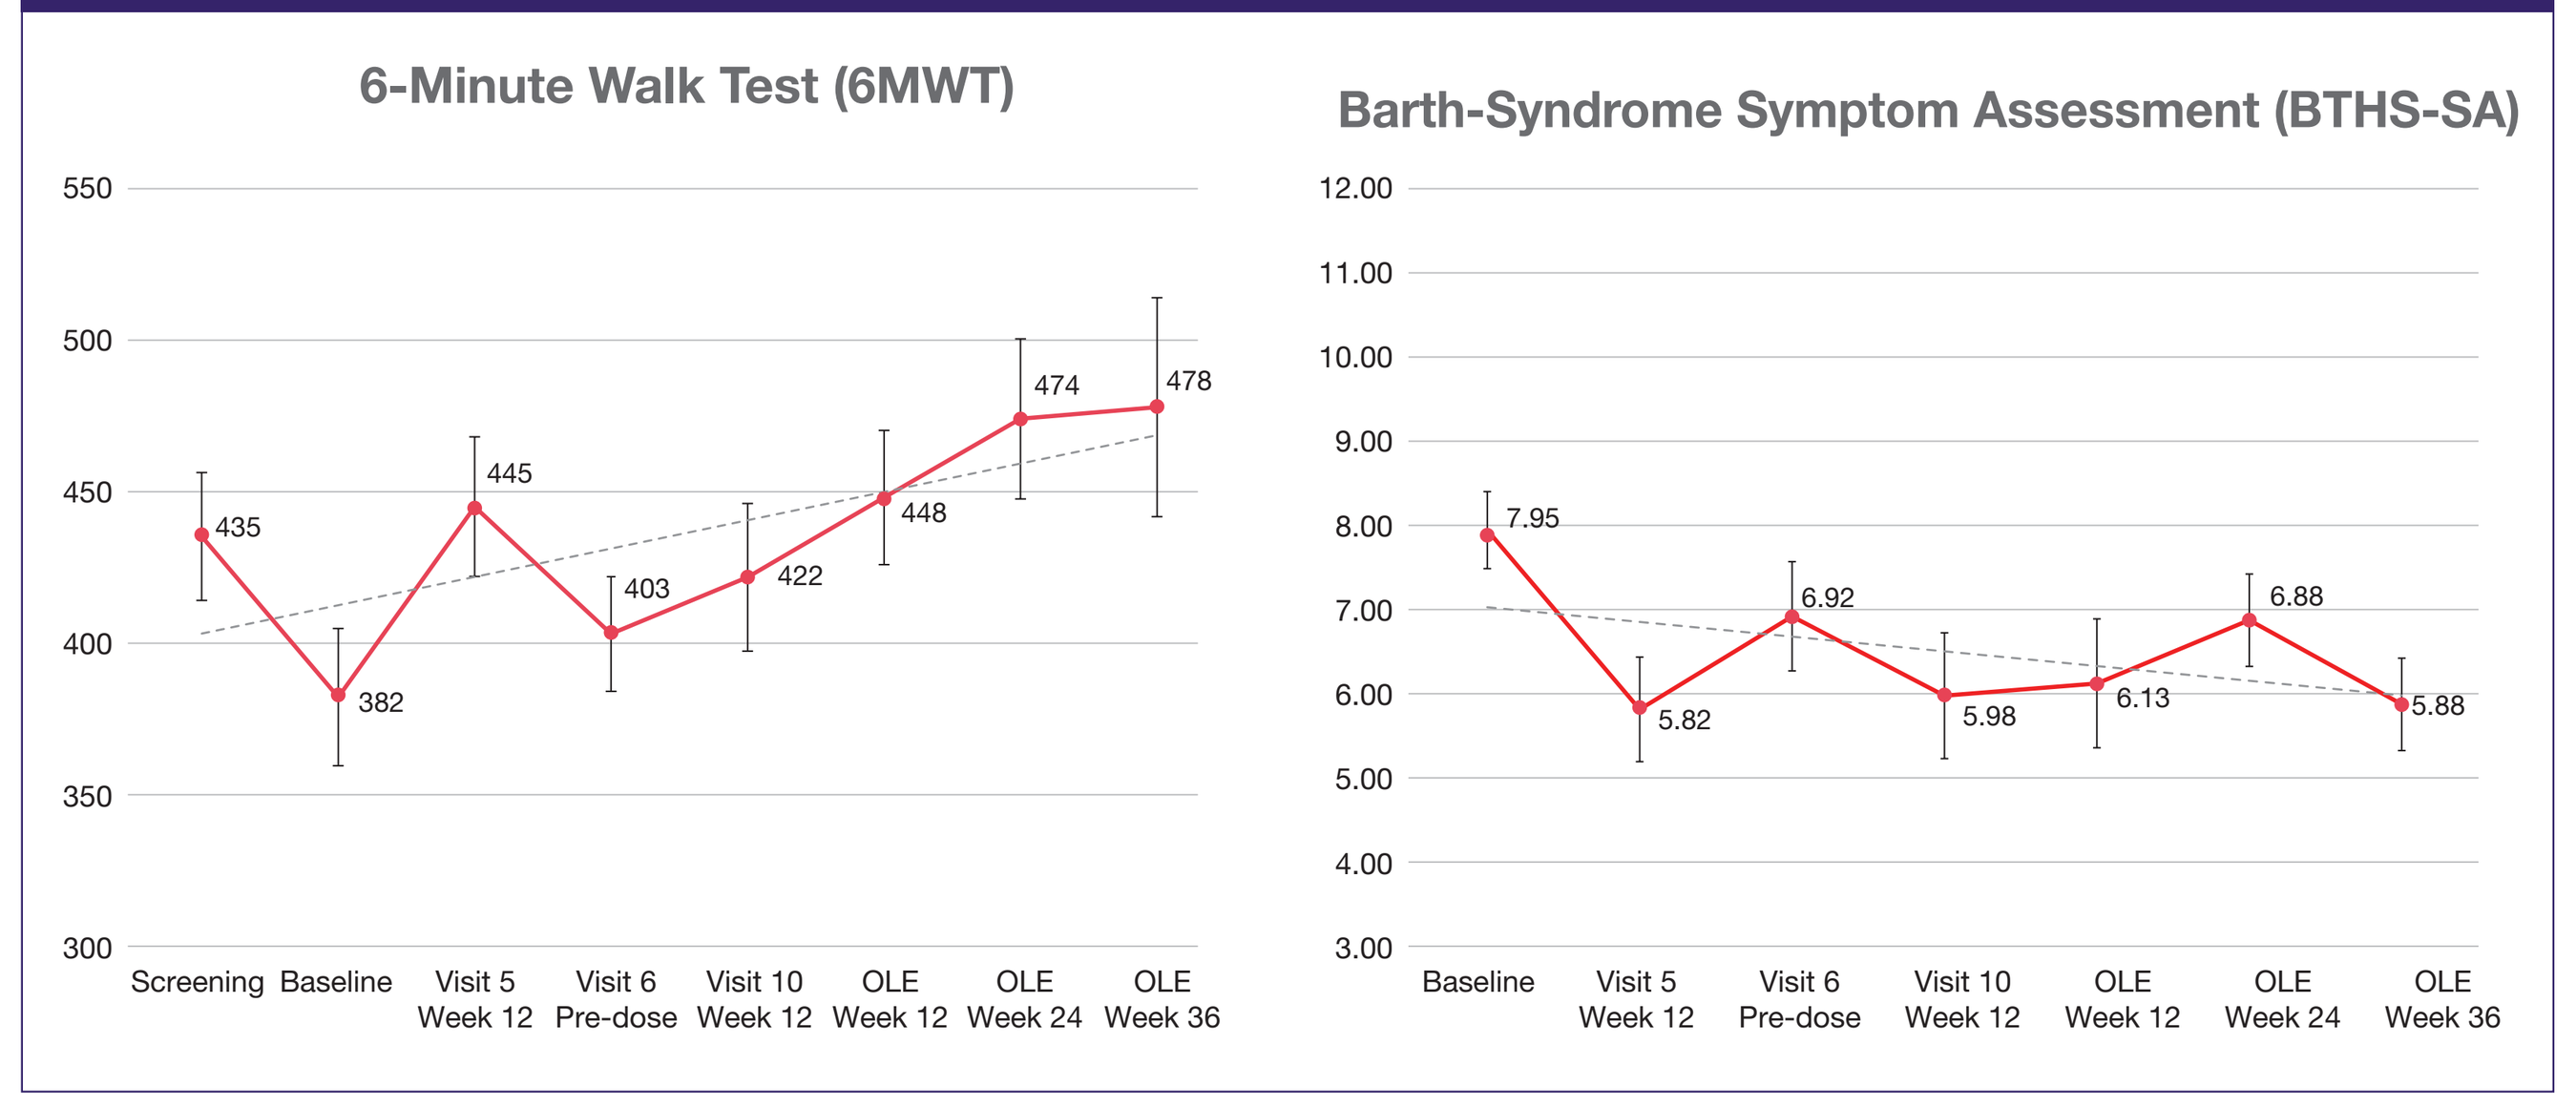
6-Minute Walk Test (6MWT)
550 500 450 400 350 300
435 382 445 403 422 448 474 478
Screening Baseline Visit 5 Week 12 Visit 6 Pre-dose Visit 10 Week 12 OLE Week 12 OLE Week 24 OLE Week 36
Barth-Syndrome Symptom Assessment (BTHS-SA)
12.00 11.00 10.00 9.00 8.00 7.00 6.00 5.00 4.00 3.00
7.95 5.82 6.92 5.98 6.13 6.88 5.88
Baseline Visit 5 Week 12 Visit 6 Pre-dose Visit 10 Week 12 OLE Week 12 OLE Week 24 OLE Week 36

Cardiac Findings

- The slope of change from baseline to OLE week 36 demonstrated increases in indexed stroke volume (p=0.0001).

Figure 3. 3D Left Ventricular (LV) Stroke Volume (Indexed to Baseline BSA)

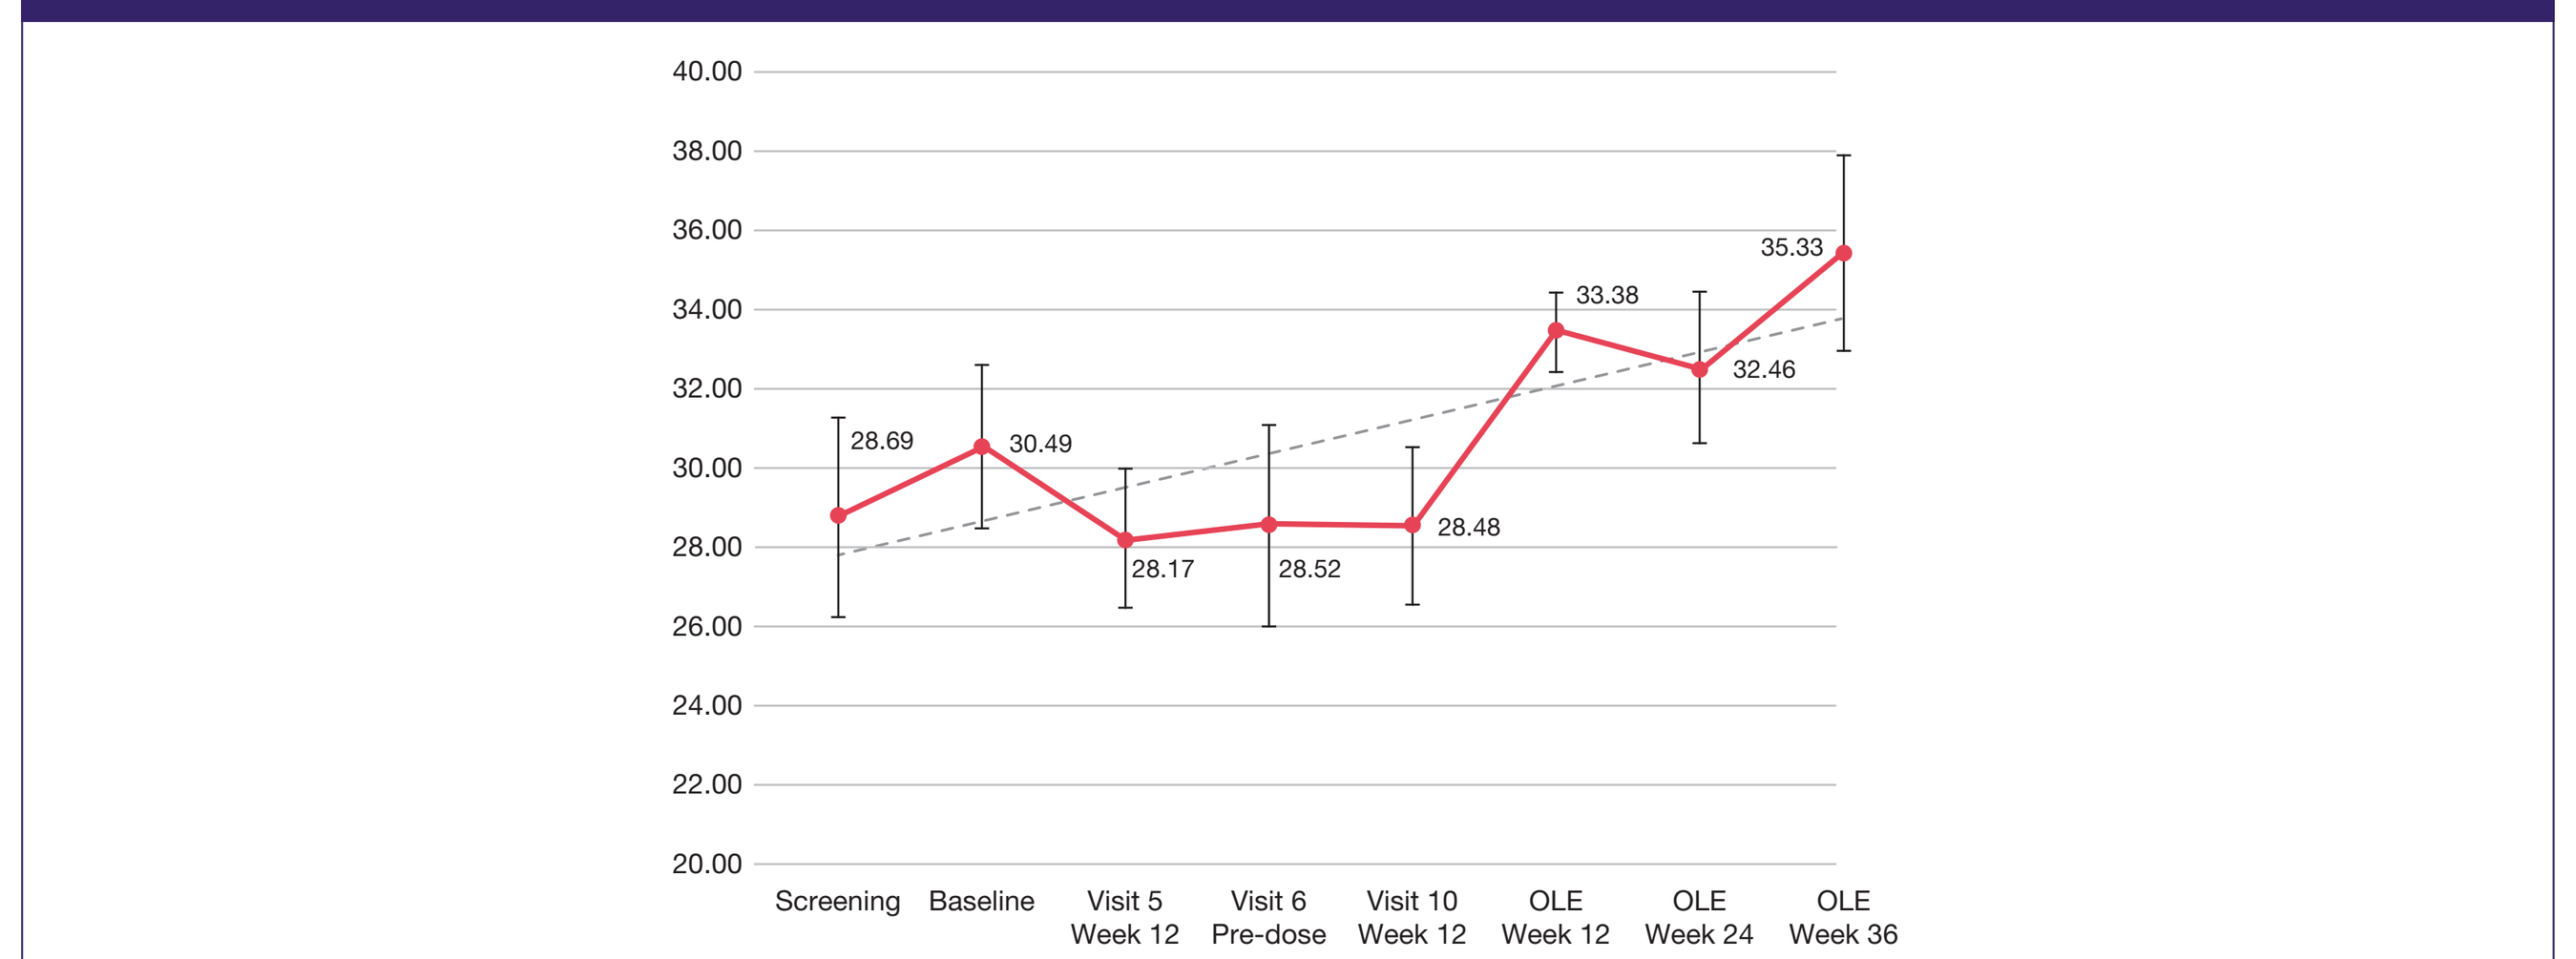
40.00 38.00 36.00 34.00 32.00 30.00 28.00 26.00 24.00 22.00 20.00
28.69 30.49 28.17 28.52 28.48 33.38 32.46 35.33
Screening Baseline Visit 5 Week 12 Visit 6 Pre-dose Visit 10 Week 12 OLE Week 12 OLE Week 24 OLE Week 36

- Correlation of change in stroke volume with changes in functional and patient-reported outcomes demonstrated a trend towards improvement in response to prolonged elamipretide treatment

Table 2. Correlation of Change in End-Systolic Volume with Changes in Functional and PRO Assessments at OLE Week 36 (n=8)

Spearman Correlation Coefficient	
Parameter	**Change (p-value)**
6MWT (meters)	0.42857 (0.2894)
HHD (newtons)	0.80952 **(0.0149)**
SWAY Balance Score	-0.04762 (0.9108)
5XSST (seconds)	-0.76190 **(0.0280)**
BTHS-SA Total Fatigue Score	-0.40476 (0.3199)
CGIS Q1	-0.38311 (0.3489)
PGIS Q1	-0.45644 (0.2556)
PROMIS Fatigue T-Score	-0.16667 (0.6932)

- Overall increase from baseline in LV end-diastolic and end-systolic volume indexed to BSA up to week 36 OLE were observed, suggesting cardiac reverse remodeling in response to prolonged elamipretide treatment
 - For LV end-diastolic volume, there was a statistically significant change from baseline (p=0.0004)
 - For LV end-systolic volume, there was a statistically significant change from baseline (p=0.0098)

Figure 4. 3D LV End Diastolic and End Systolic Volume (Indexed)

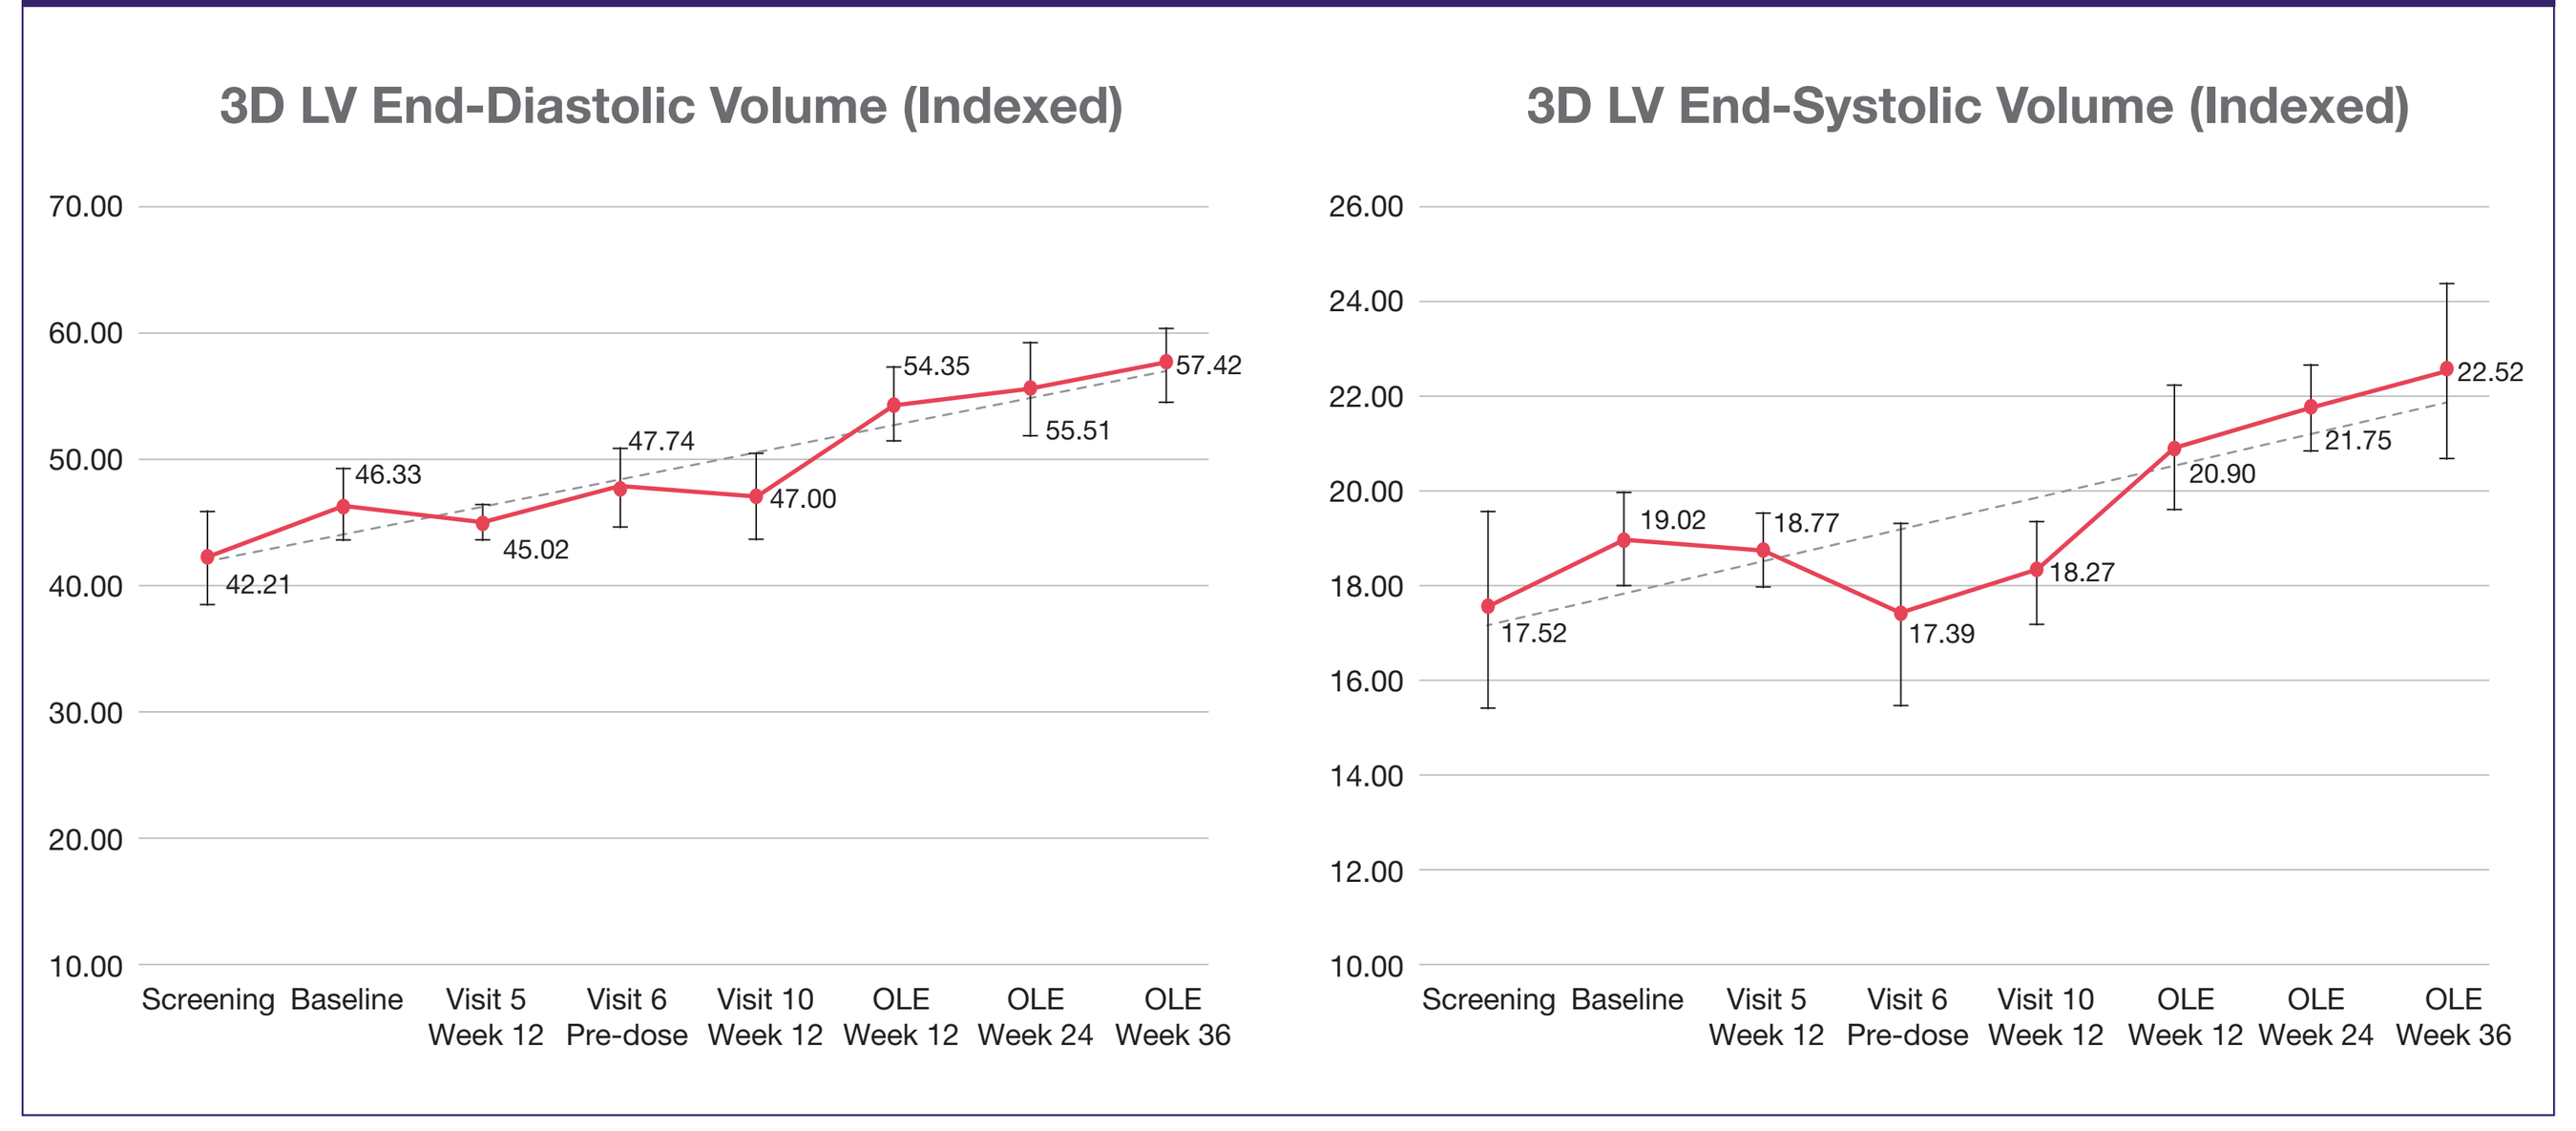
3D LV End-Diastolic Volume (Indexed)
70.00 60.00 50.00 40.00 30.00 20.00 10.00
42.21 46.33 45.02 47.74 47.00 54.35 55.51 57.42
3D LV End-Systolic Volume (Indexed)
26.00 24.00 22.00 20.00 18.00 16.00 14.00 12.00 10.00
17.52 19.02 18.77 17.39 18.27 20.90 21.75 22.52
Screening Baseline Visit 5 Week 12 Visit 6 Pre-dose Visit 10 Week 12 OLE Week 12 OLE Week 24 OLE Week 36

- LV mass indexed to BSA showed an overall increase from baseline up to OLE Week 36, indicating improvement in cardiac structure over time in response to prolonged elamipretide treatment

Table 3. Change from Baseline to OLE Week 36 in Cardiac Parameters

Endpoint	Baseline: Predose Actual Value Mean (SD)	Change from Baseline to OLE Week 36 Mean (SD, p-value)
2D Echo Peak E/Peak A Wave	1.800 (0.7869)	0.144 (0.6550, p=0.5546)
Global Longitudinal Strain (Triplane)	-19.82 (1.693)	0.92 (1.854, p=0.2010)
LV Mass (M-mode) BSA Z-Score	 82.0 (17.30) -0.371 (0.8964)	 12.0 (19.56, p=0.1547) 0.657 (1.1060, p=0.1673)
Non-Compaction Ratio at LV Apex at End-Diastole from 2D Echo (Chin Method)	0.326 (0.0964)	0.039 (0.0770, p=0.1976)
Non-Compaction Ratio at LV Apex at End-Systole from 2D Echo (Jenni Method)	1.427 (0.5580)	-0.289 (0.4537, p=0.1149)
Tricuspid Valve Regurgitation from 2D Echo	1.2 (0.63)	-0.4 (0.52, p=0.0796)

Safety and Tolerability

- There were 91 TEAEs reported, with 1 serious AE deemed not related to elamipretide; injection site reactions occurred in 100% of patients while on elamipretide

Table 4. Treatment-Emergent Adverse Events

System Organ Class Preferred Term	Blinded Trial Adverse Events (N=12)		OLE Adverse Events (N=10)
	Elamipretide, n (%)	**Placebo, n (%)**	**Elamipretide, n (%)**
General disorders			
Aphthous ulcer	0	2 (16.7)	0
Gastrointestinal disorders			
Nausea	0	0	2 (20)
General disorders and administrative site conditions			
Injection site erythema	12 (100)	3 (25)	8 (80)
Injection site pain	9 (75.0)	4 (33.3)	7 (70)
Injection site induration	8 (66.7)	2 (16.7)	5 (50)
Injection site pruritus	8 (66.7)	2 (16.7)	7 (70)
Injection site bruising	3 (25.0)	0	0
Injection site urticaria	3 (25.0)	0	0
Medical device site irritation	2 (16.7)	1 (8.3)	0
Infections and infestations			
Bronchitis	2 (16.7)	1 (8.3)	0
Viral upper respiratory tract infection	1 (8.3)	2 (16.7)	0
Pharyngitis streptococcal	1 (8.3)	2 (16.7)	0
Ear infection	0	0	2 (20)
Gingivitis	0	0	2 (20)
Injury, poisoning and procedural complications			
Ligament sprain	2 (16.7)	1 (8.3)	0
Joint dislocation	0	0	2 (20)*
Muscle strain	0	0	2 (20)
Nervous system disorders			
Headache	1 (8.3)	3 (25.0)	3 (30)
Dizziness	0	0	4 (40)
Musculoskeletal and connective tissue disorders			
Arthralgia	0	0	2 (20)
Pain in extremity	0	0	2 (20)
Respiratory, thoracic and mediastinal disorders			
Cough	0	0	2 (20)
Oropharyngeal pain	0	0	2 (20)

*Joint dislocation was reported as severe, all other adverse events in the OLE were mild or moderate in intensity.

CONCLUSIONS

- TAZPOWER is the first clinical trial to evaluate the tolerability and efficacy of a potential therapeutic agent in patients with BTHS
- Blinded Phase of the TAZPOWER Trial
 - Statistical significance was not achieved in the ITT population on the primary endpoints; elamipretide provided clinically meaningful improvements in individual functional and PROs
 - Elamipretide was generally well tolerated; most adverse events were mild to moderate in severity, with the most commonly reported adverse events including injection site reactions
- Open-Label Extension Phase of the TAZPOWER Trial
 - At 36-week OLE, elamipretide therapy was associated with improvements in 6MWT and BTHS-SA Total Fatigue Score
 - Safety and tolerability of elamipretide was consistent with blinded phase observations
- Cardiac Findings during Open-Label Extension
 - Cardiac findings during 36-week OLE showed increased LV end-diastolic and systolic volumes, leading to higher stroke volumes

Disclosures: The authors declare no conflict of interest.
Study Funding: Trial funded by Stealth BioTherapeutics, Newton, MA
Medical writing assistance was provided by James A. Shiffer, RPh, Write On Time Medical Communications, LLC

To view this poster on the Stealth BioTherapeutics website, snap a photo of this QR code with your smart phone (QR reader required).

Stealth BIOTHERAPEUTICS